Chimeric and Rationally Designed Compact Promoters for Cardiomyocyte-Specific Gene Expression

Prasad R. Konkalmatt , Chris A. Reid, Melissa Van Pell, Samantha Jones, Ze Cheng, Joe Woods, Anastasiia Budan, Alex Aycinena, Mo Mandegar, Kathy Ivey,
Tenaya Therapeutics, Inc. South San Francisco, CA – 94080, USA.

INTRODUCTION

Tenaya Therapeutics has established integrated internal capabilities to broadly enable modality agnostic target validation and the identification, selection and optimization of capsids and components best suited to constructing and manufacturing adeno-associated virus (AAV)-based genetic medicines intended to target the underlying cause of cardiac diseases.

AAV vectors have emerged as preferred gene delivery vehicles for numerous human gene therapy applications. AAVs demonstrate the ability to transduce various organs and cell types, including cardiomyocytes (CMs), leading to sustained gene expression. This characteristic makes them well-suited for treating genetic disorders that necessitate prolonged therapeutic gene expression. While the broad tissue tropism exhibited by AAV serotypes is advantageous for targeting multiple organs, it can also lead to off-target expression if the AAV reaches unintended organs and cell types potentially resulting in adverse clinical outcomes. Selection of promoters tailored to the target tissue and cell types is crucial for achieving the desired expression profile in gene therapy applications.

One of the limitations with AAVs is their small packaging capacity. The optimal AAV genome size is around 4.7 kb, which restricts the size of the gene of interest that can be delivered. The development of shorter CM-specific promoters represents a promising strategy to enhance the specificity, efficacy, and safety of AAV-based gene therapy. Additionally, it enables AAV gene therapies for larger genes of interest and gene editing without surpassing the ideal packaging capacity of ~4.7 kb

OBJECTIVE

Objective of the project is to develop compact cardiomyocyte (CM) specific promoters that maintain promoter activity and specificity, thus reducing the overall size of the promoter to accommodate larger therapeutic payloads.

MATERIALS AND METHODS

Chimeric CM library preparation and screening: To develop a shorter CM-specific promoter, we employed a novel approach to create a library of 250 bp chimeric promoters and screened them for cardiac-specific gene expression. These chimeric promoters were constructed using the natural 500 bp proximal promoter regions of five genes, *MYBPC3, MYH6, MYH7, MYL3* and *TNNT2*, that are preferentially expressed in the adult CM. The 500 bp promoter sequences were digitally digested into 50bp fragments and shuffled in silico using a custom-built python code. This process resulted in the generation of 2,500 unique chimeric promoters, each 250 bp in length. These novel promoters were synthesized as pooled oligos and cloned upstream of a GFP reporter transgene. Subsequently, ~1,000 unique chimeric promoters were isolated and assessed for their efficiency in driving GFP expression in human iPSC-CMs following transient transfection.

In vivo evaluation of chimeric CM promoters:. Each chimeric CM promoter-GFP constructs was assigned with 3 unique molecular barcodes located between the GFP open reading frame (ORF) and the bovine growth hormone (bGH) poly-adenylation signal (PolyA). All promoter constructs were packaged into AAV9 capsid by triple transfection method and purified by Iodixanol gradient centrifugation. The AAVs were pooled maintaining the same number of viral genome copies for each promoter construct. The AAV pool was injected at the dose of, 1.5E+13 vg/kg body weight to cynomolgus monkeys (n=2), and 3E+13 vg/kg body weight to C57Bl/6 mice (n=4). Heart and liver tissue samples were collected at 4-week post-injection from cynomolgus monkeys, and 3-week post-injection from mice. The barcoded regions were subjected to NGS analyses following PCR amplification of the RNA and DNA prepared from the tissues. The promoter strengths were determined by comparing the relative number of the transcripts via NGS analyses of the barcodes associated with each promoter. NGS read count of each barcode in each sample was normalized to the total number of reads in the sample, the abundance of the barcode in the initial virus pool, and the measurement of reference cassette in the pool. The counts for all 3 barcodes associated with each promoter construct were averaged. Finally, the RNA counts for construct was normalized to the corresponding viral DNA count in the same sample to generate the final performance score of each promoter.

In silico analyses of promoter sequences: in silico analyses of the promoters for transcription factor binding sites, and TATA box and TTS elements was carried out using the online software tools https://jaspar.genereg.net and https://www.fruitfly.org/cgi-bin/seq_tools/promoter.pl respectively.

MATERIALS AND METHODS, cont'd

In vitro evaluation of 6C2 and modified CM promoters: iPSC-CM cells were transfected with the indicated promoter-GFP constructs directing the expression of GFP. Six days after, the cells were fixed with 4% PFA and the GFP fluorescence was measured using Cytation 5 imaging reader (Molecular Devices). Relative promoter strengths were determined by comparing the fluorescence levels produced by different promoter-GFP constructs. The promoter strengths were normalized to 400 bp human TNNT2 promoter.

In vivo evaluation of 6C2 and modified CM promoters: The promoter constructs were packaged into AAV9 capsid by triple transfection method and purified by Iodixanol gradient centrifugation. The AAVs were injected into C57Bl/6 mice retro-orbitally at the dose of 1E+13 vg/kg body weight (n=4). Heart and liver tissues were collected at 4-week post-injection. Protein extracts from the tissue samples were electrophoresed under reducing conditions on 4–12% gradient polyacrylamide gels and transferred to nitrocellulose membranes. The membranes were probed with rabbit anti-GFP antibodies (ab-13970, Abcam), followed by Chicken anti GAPDH antibodies (GTX85118, GeneTex). The intensities of the GFP signals were normalized to the corresponding GAPDH signals on the same membrane. The results were expressed as fold difference compared to the intensities produced from 400 bp human TNNT2 promoter-GFP construct.

RESULTS

The chimeric CM promoter library showed even distribution of parental promoter sequences

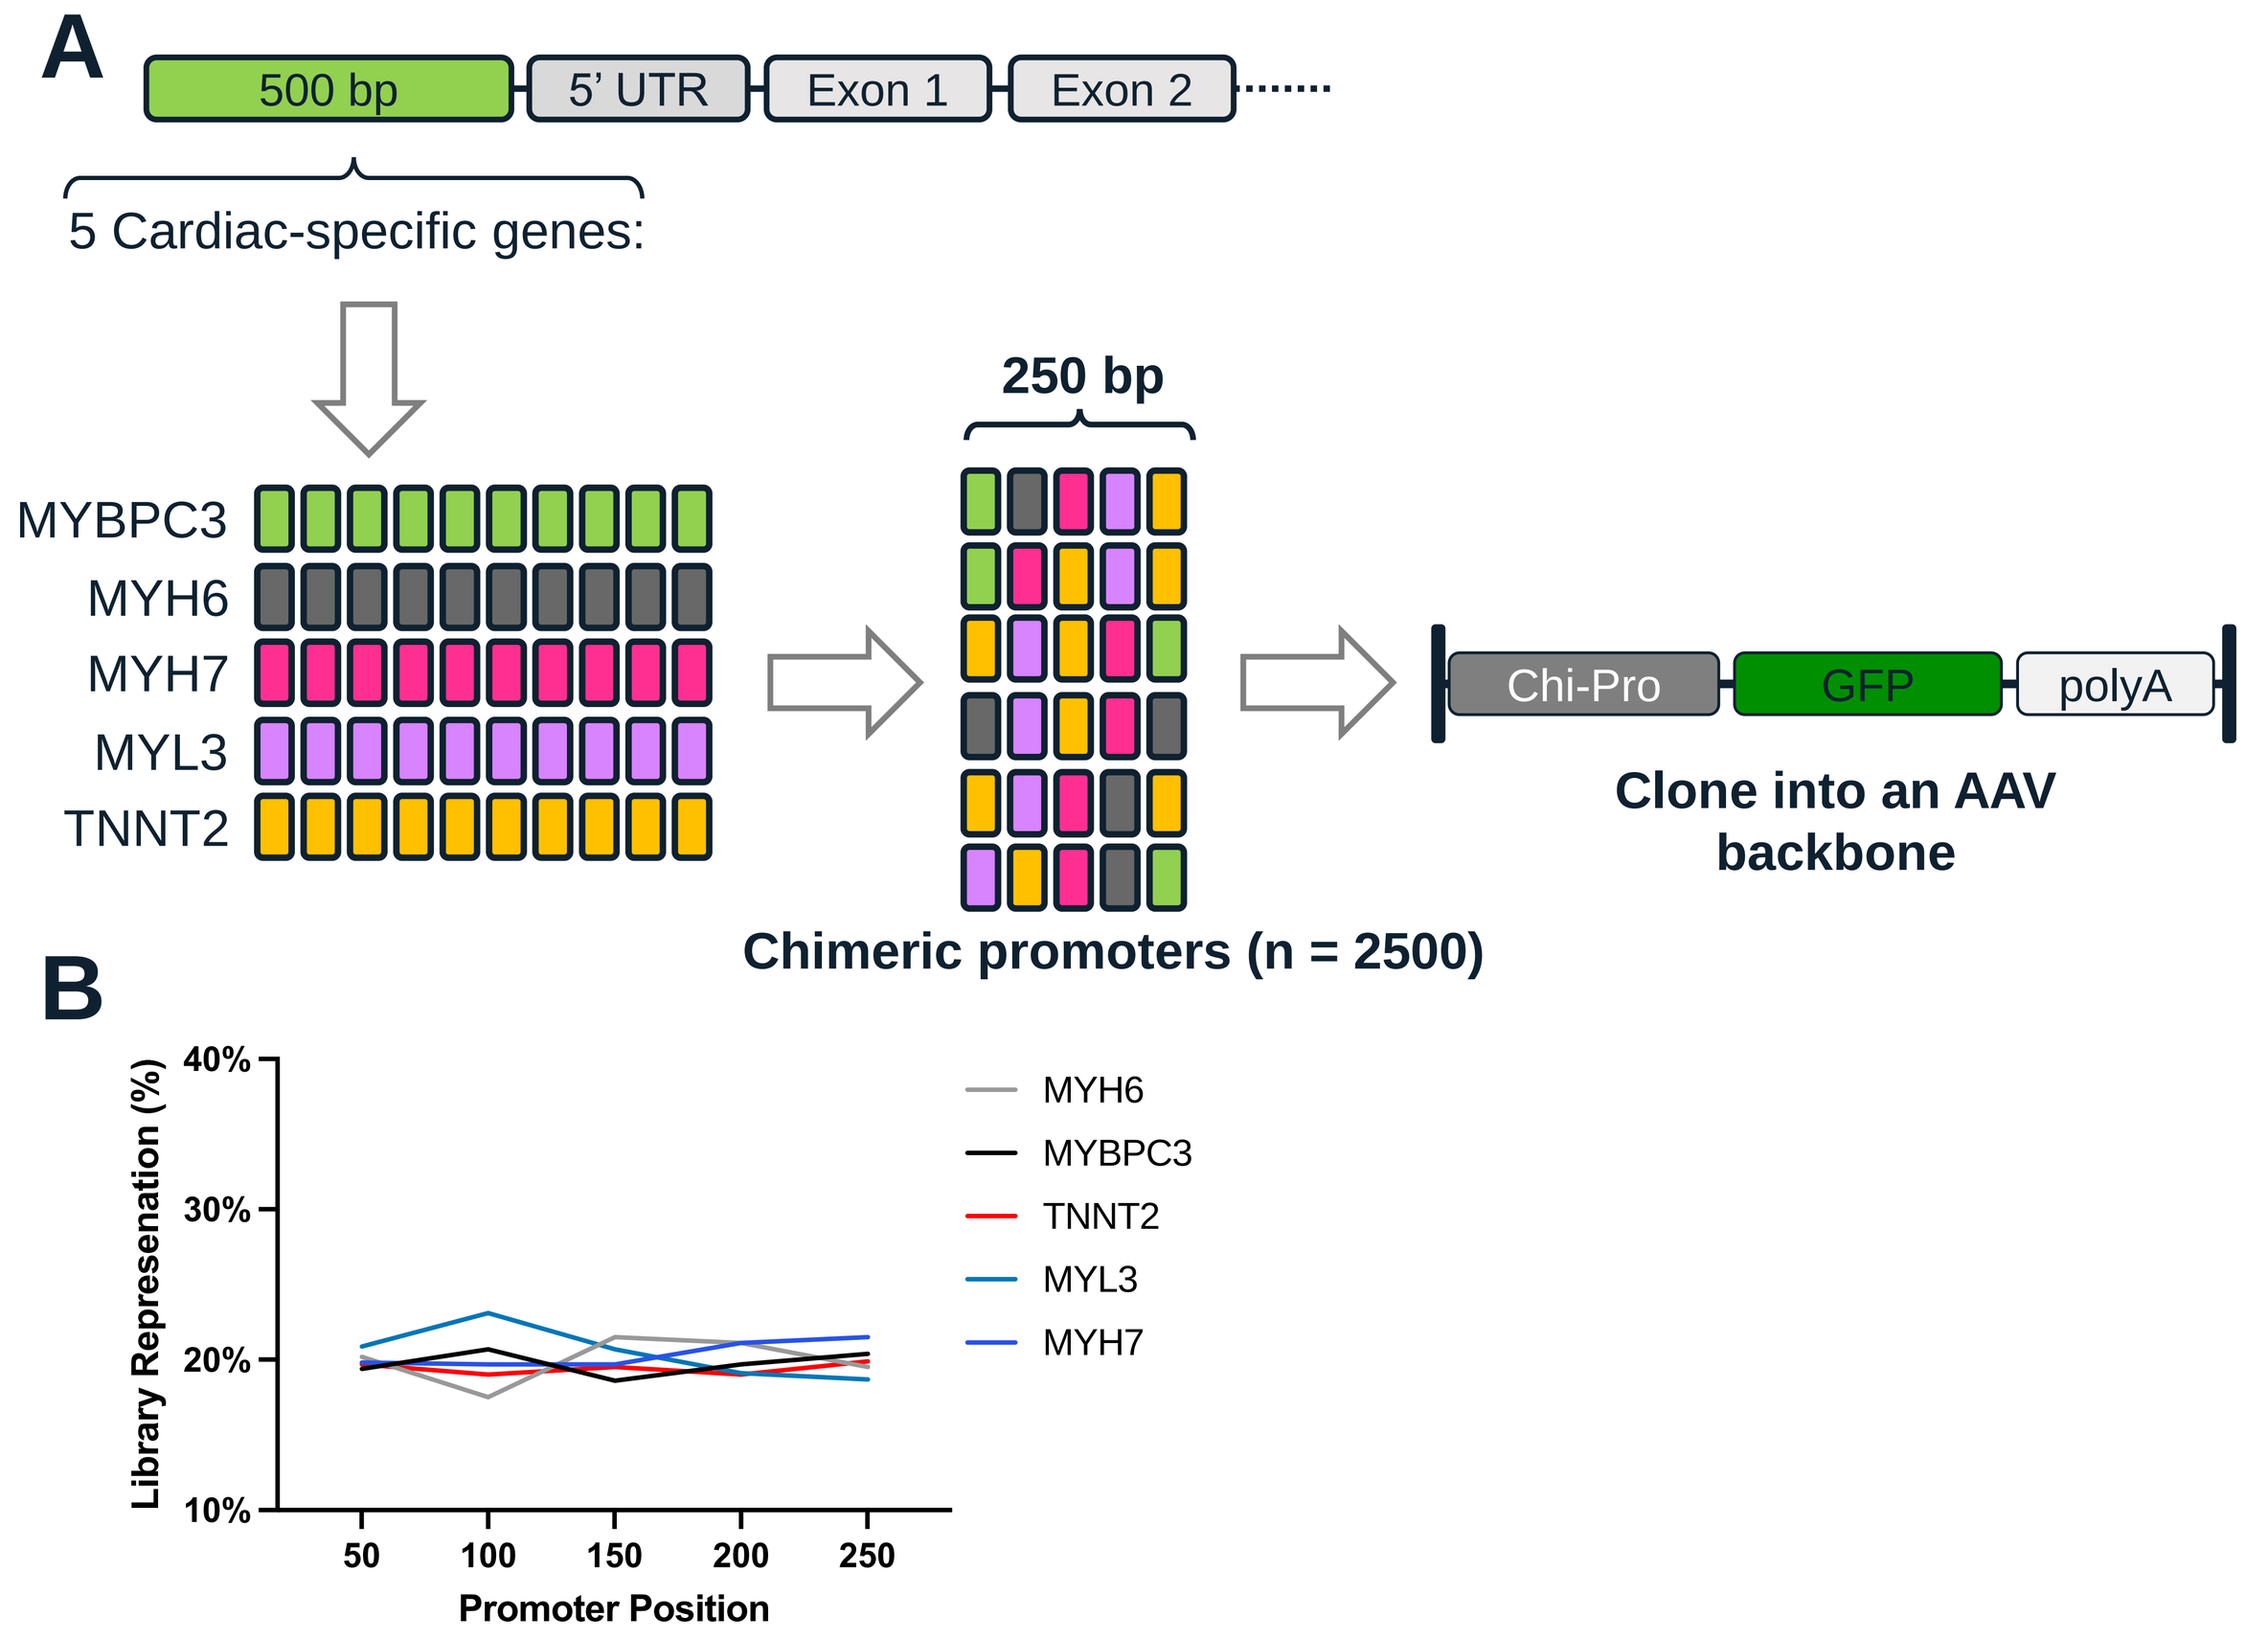
A
500 bp
5' UTR
Exon 1
Exon 2
5 Cardiac-specific genes:
MYBPC3
MYH6
MYH7
MYL3
TNNT2
250 bp
Chimeric promoters (n = 2500)
Chi-Pro
GFP
polyA
Clone into an AAV backbone
B
Library Represenation (%)
Promoter Position
MYH6
MYBPC3
TNNT2
MYL3
MYH7

Figure 1. The 500 bp proximal promoter sequences of five genes, MYBPC3, MYH6, MYH7, MYL3 and TNNT2, were digitally digested into 50bp fragments and shuffled in silico using a custom-built python code. This process resulted in the generation of 2,500 unique chimeric promoters, each 250 bp in length. These novel promoters were synthesized as pooled oligos and cloned upstream of a GFP reporter transgene. B) Graph showing each of the 5 natural promoter sequence is represented evenly at each of the 50 bp location in the chimeric CM library

In vitro screening of the chimeric CM promoter library identified several variants exhibiting promoter activity in iPSC-CM cells

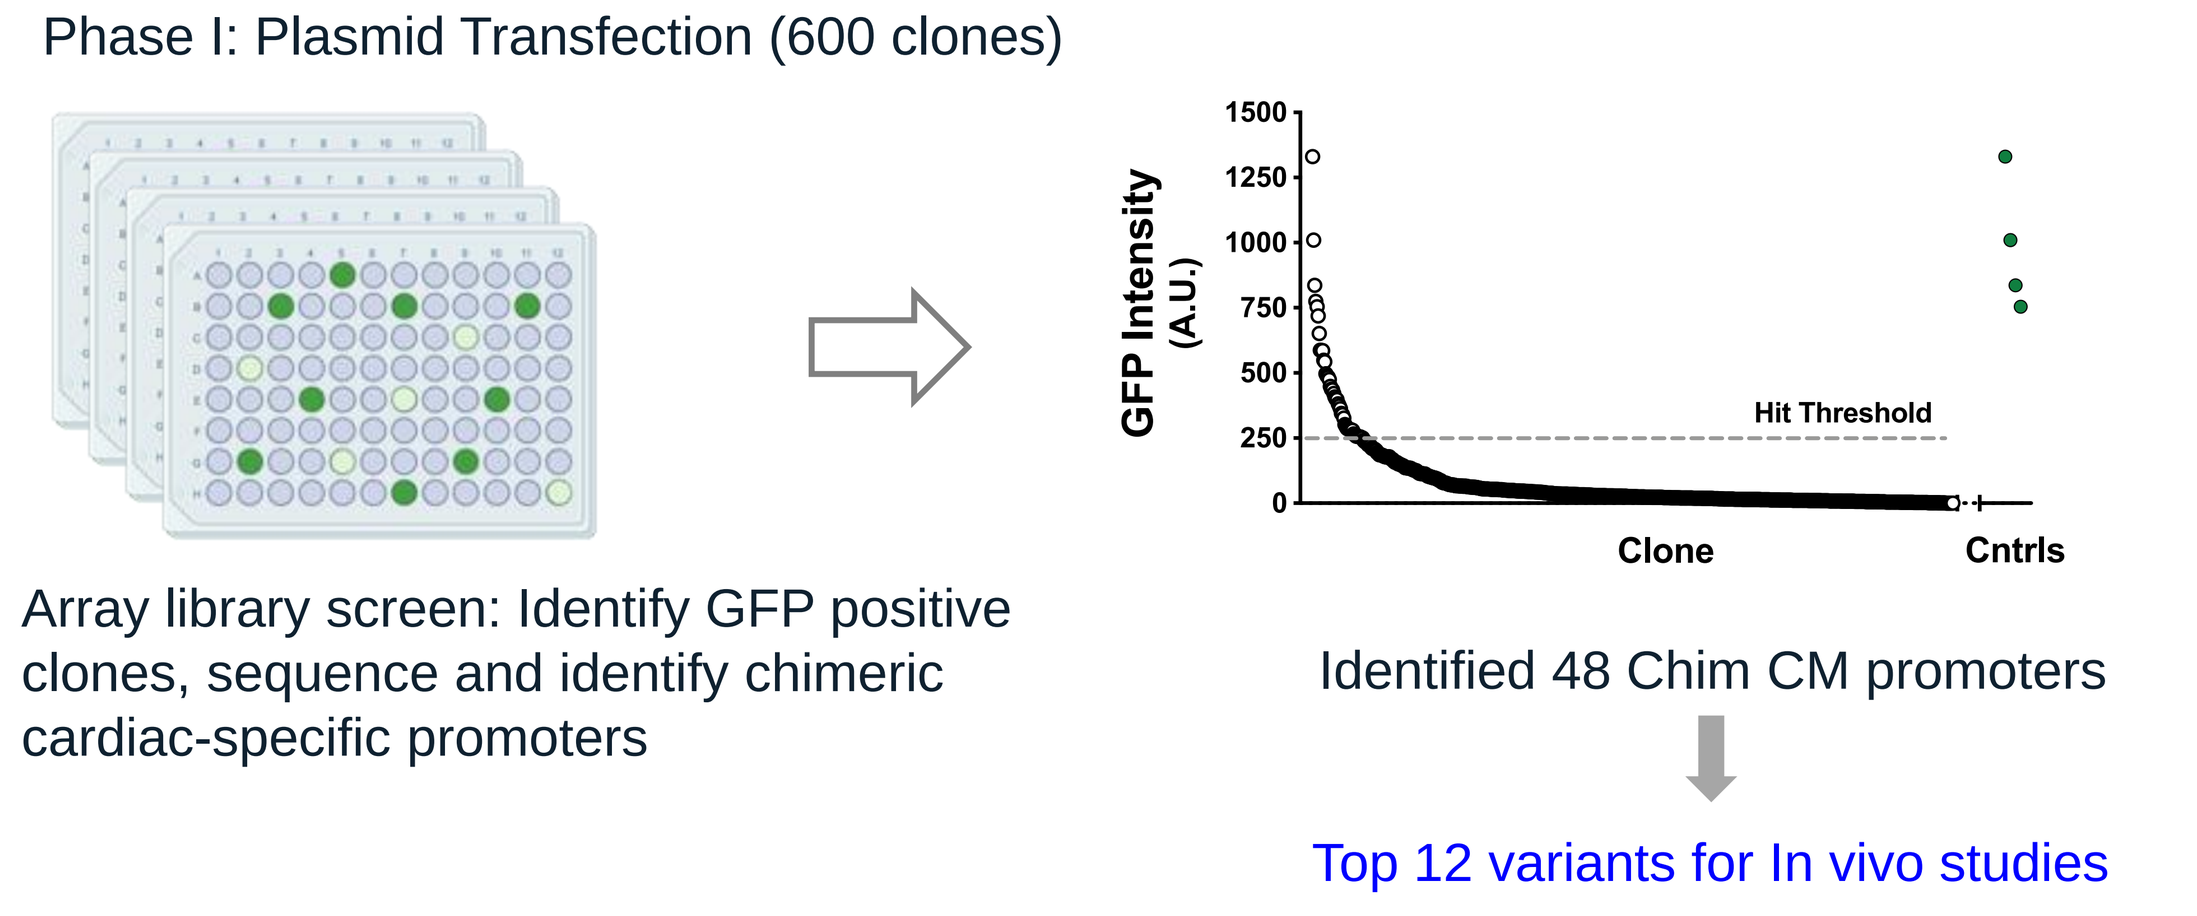
Phase I: Plasmid Transfection (600 clones)
Array library screen: Identify GFP positive clones, sequence and identify chimeric cardiac-specific promoters
GFP Intensity (A.U.)
Hit Threshold
Clone
Cntrls
Identified 48 Chim CM promoters
Top 12 variants for In vivo studies

Figure 2. Approximately 1,000 unique chimeric promoters from the library were isolated and assessed for their efficiency in driving GFP expression in human iPSC-CMs following transient transfection. The top 12 variants were chosen for in vivo evaluation in cynomolgus monkeys and mice.

RESULTS, cont'd

In vivo evaluation of the selected chimeric CM promoters identified a variant 6C2 exhibiting CM specific gene expression

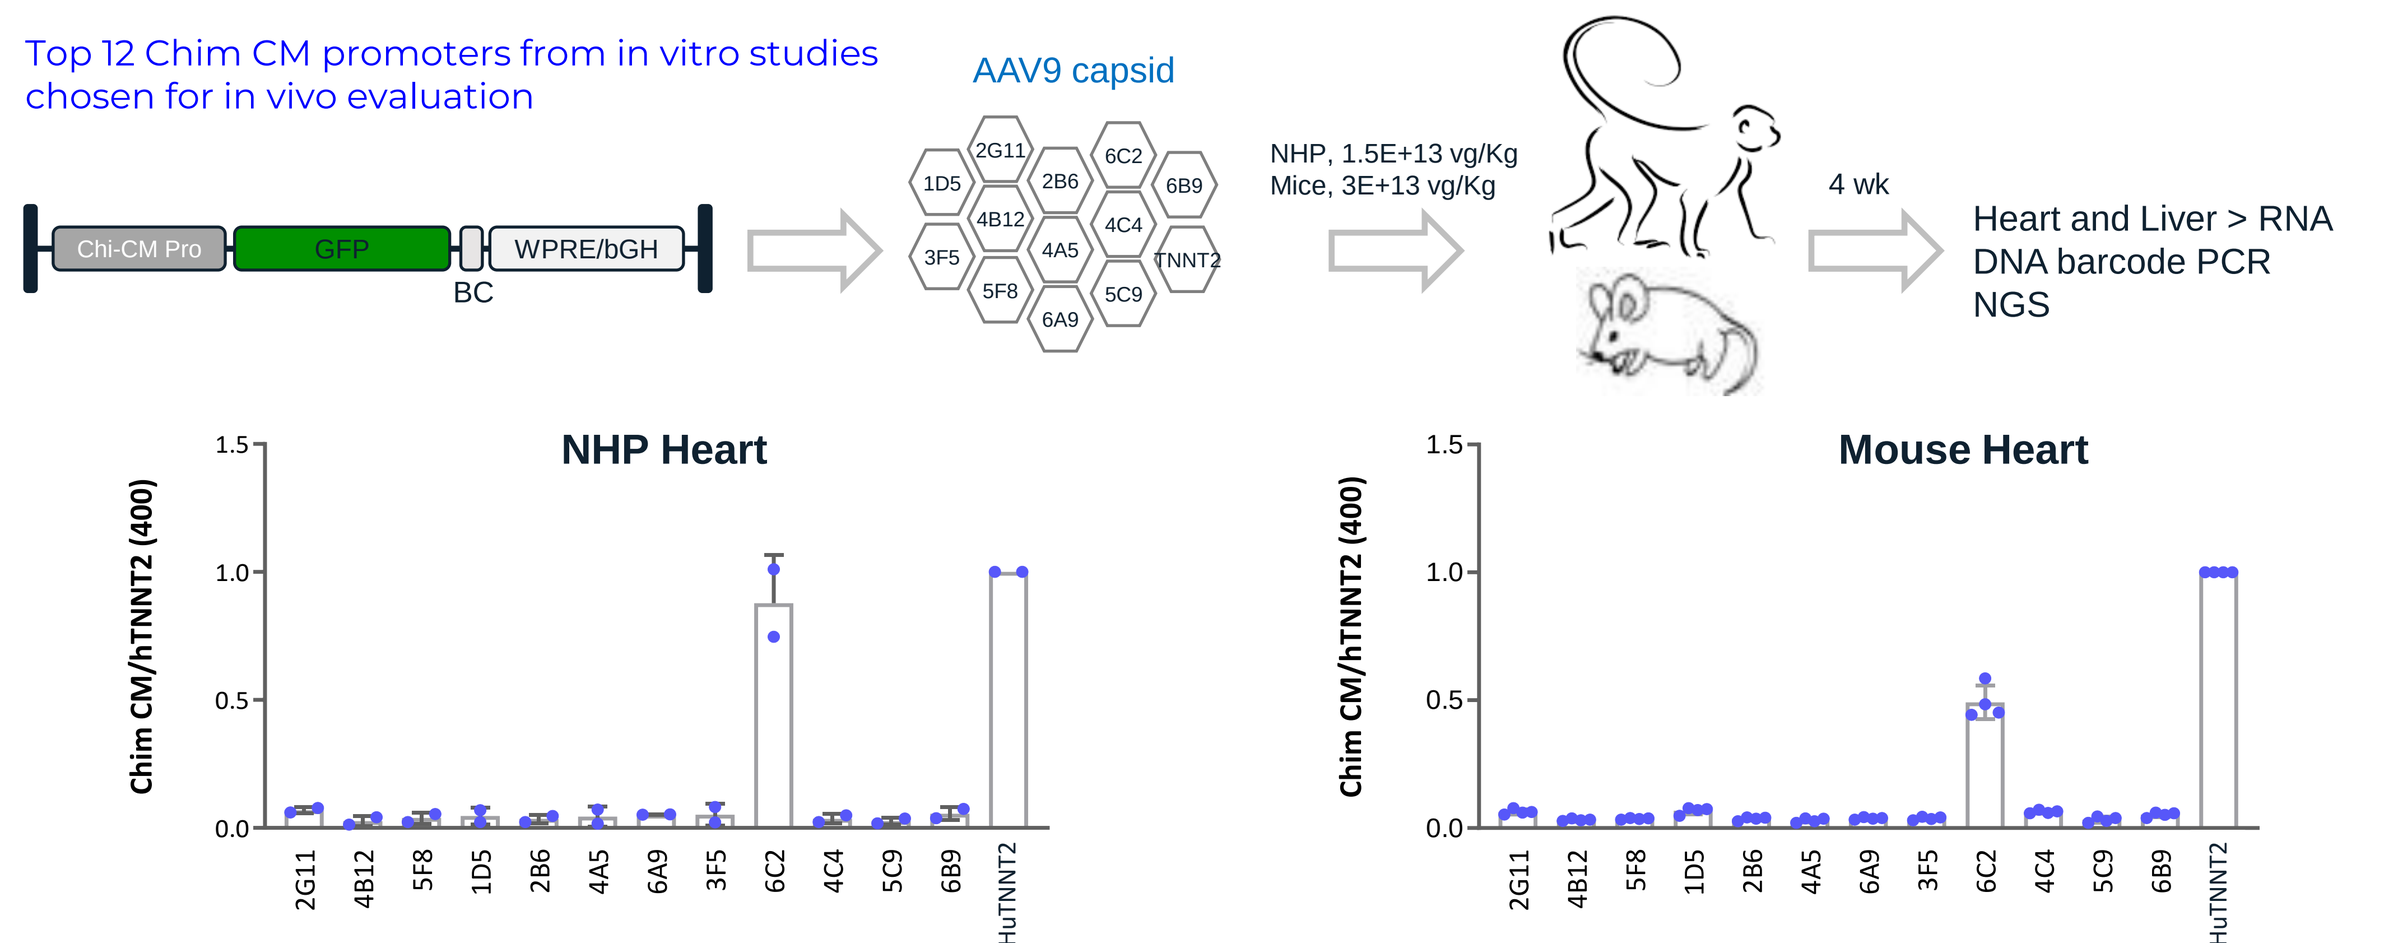
Top 12 Chim CM promoters from in vitro studies chosen for in vivo evaluation
AAV9 capsid
Chi-CM Pro
GFP
BC
WPRE/bGH
NHP, 1.5E+13 vg/Kg
Mice, 3E+13 vg/Kg
4 wk
Heart and Liver > RNA DNA barcode PCR NGS
NHP Heart
Mouse Heart
Chim CM/hTNNT2 (400)

Figure 3. The 12 chimeric promoters assigned with unique molecular barcodes were packaged in AAV9 capsids pooled and administered in the cynomolgus monkeys and in mice. 3 or 4 weeks after the promoter strengths were determined by NGS analyses. Out of the 12 chimeric CM promoters, one (6C2) produced gene expression levels of 0.85 ± 0.18-fold in NHP hearts, 0.5 ±0.11-fold in the mouse hearts compared to 400 bp human TNNT2 promoter.

In silico evaluation of chimeric CM promoter 6C2 revealed the composition, CM specific TSBS and locations of core promoter elements

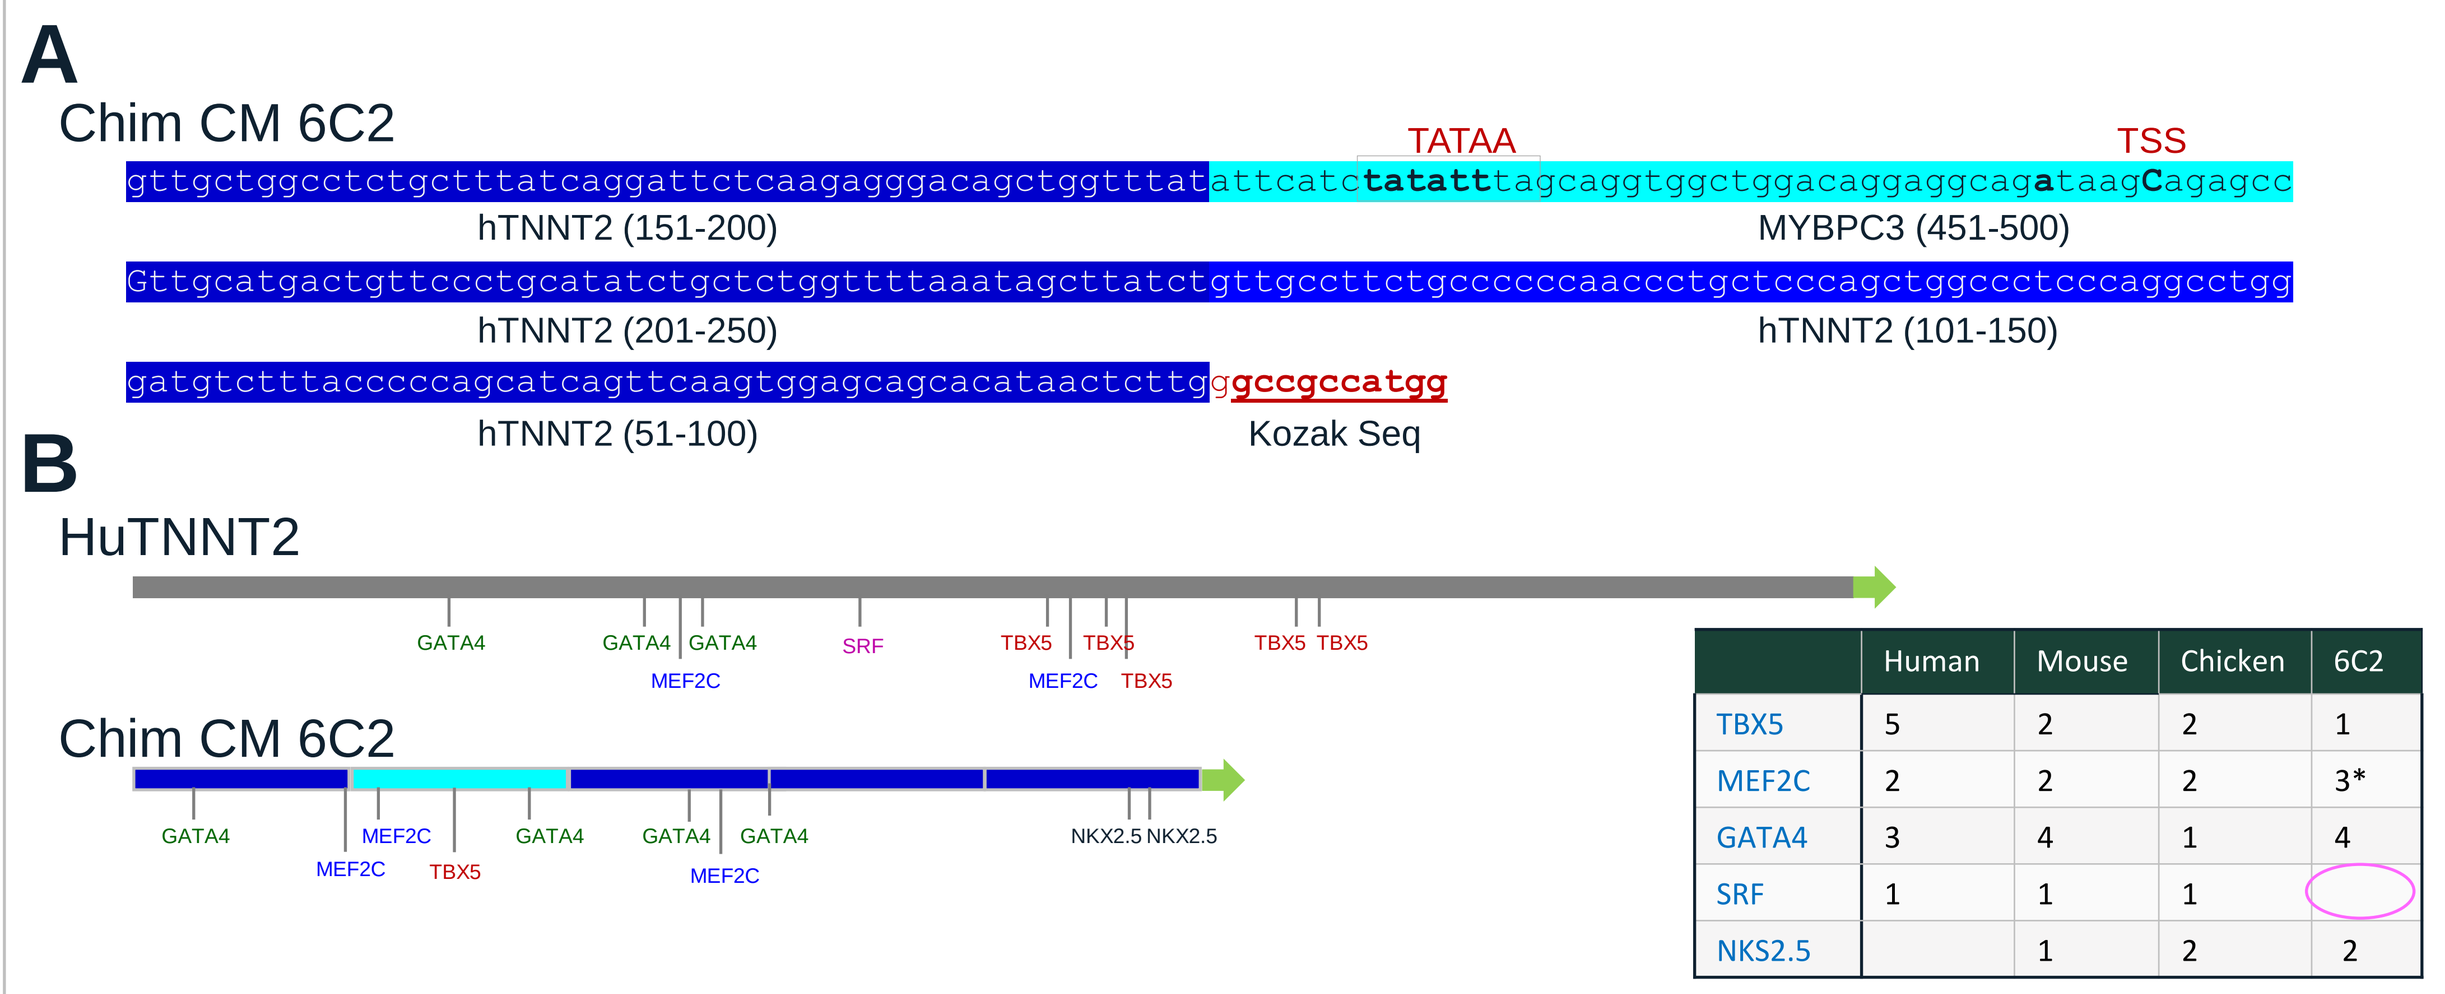
A
Chim CM 6C2
gttgctggcctctgctttatcaggattctcaagagggacagctggtttatattcatctatatttagcaggtggctggacaggaggcagataagCagagcc
hTNNT2 (151-200)
TATAA
TSS
MYBPC3 (451-500)
Gttgcatgactgttccctgcatatctgctctggttttaaatagcttatctgttgccttctgccccccaaccctgctcccagctggccctcccaggcctgg
hTNNT2 (201-250)
hTNNT2 (101-150)
gatgtctttaccccagcatcagttcaagtggagcagcacataactcttggccgccatgg
hTNNT2 (51-100)
Kozak Seq
B
HuTNNT2
Chim CM 6C2

	Human	Mouse	Chicken	6C2
TBX5	5	2	2	1
MEF2C	2	2	2	3*
GATA4	3	4	1	4
SRF	1	1	1	
NKS2.5		1	2	2

Figure 4. In silico analyses of chimeric CM promoter 6C2 showing, A) nucleotide sequence of 6C2, identity of the five 50 bp fragments and their location in the natural promoters, and the TATA box, TSS and kozak seq elements is indicated. B) Diagram showing the locations of CM specific transcription factor binding sites (TFBS) in human TNNT2 and 6C2 promoter (http://jasper.genereg.net; Relative profile threshold = 80%). Table showing the number of CM specific TFBS in human, mouse, chicken TNNT2 promoter and 6C2.

Addition of CMV core promoter at the 3' end of 6C2 enhanced its promoter strength in iPSC-CM cells

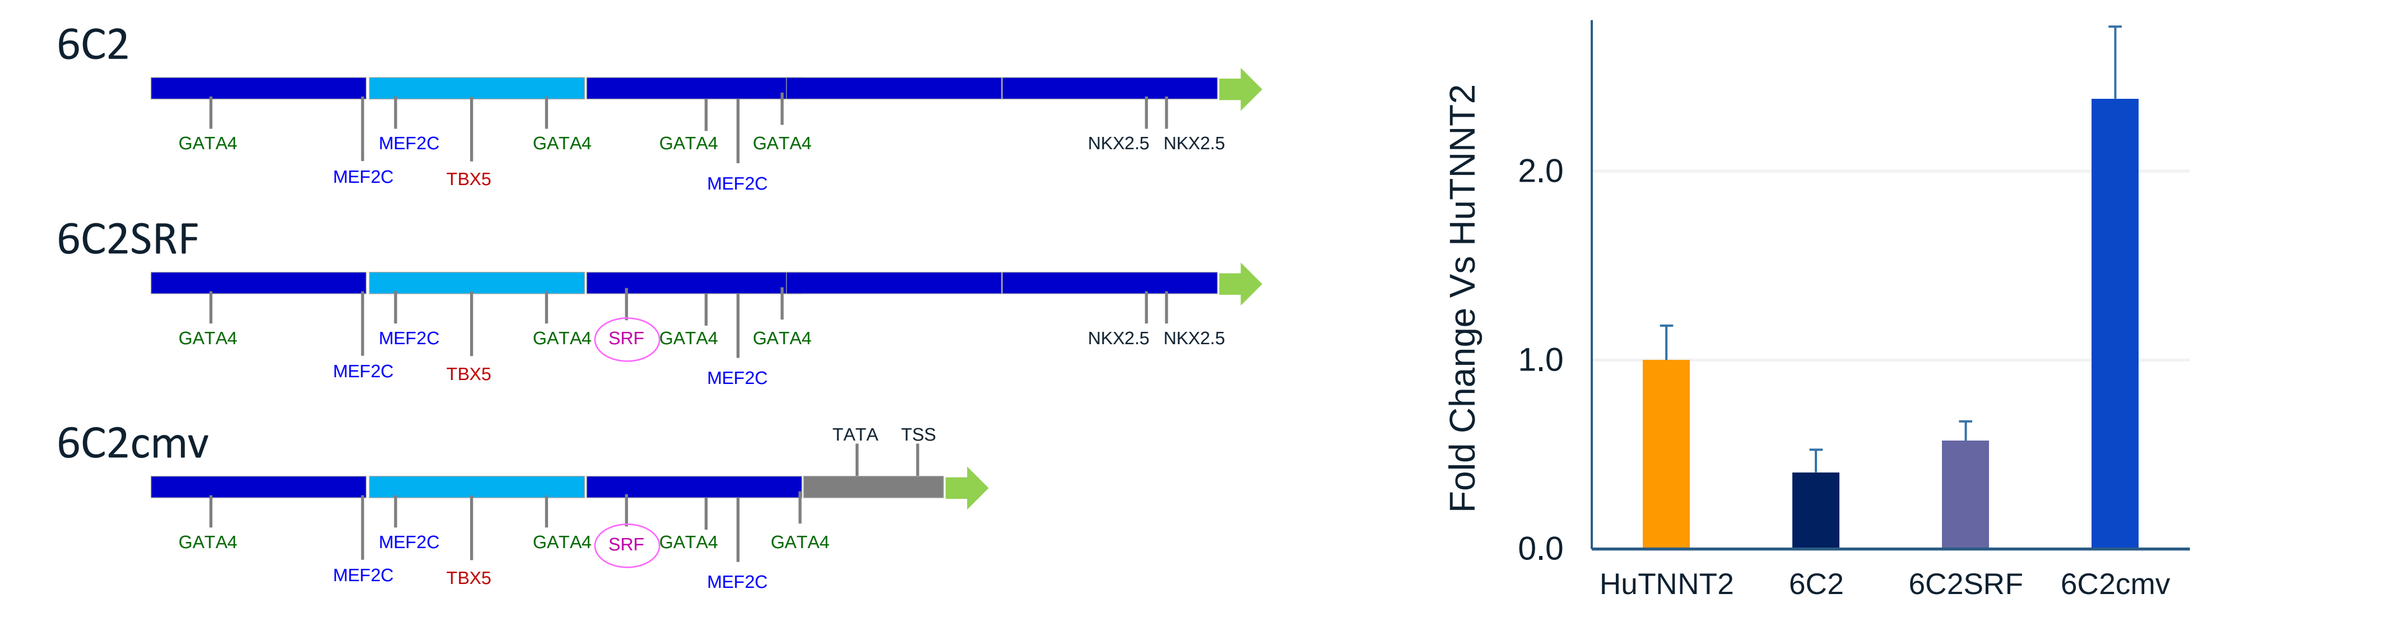
6C2
6C2SRF
6C2cmv
Fold Change Vs HuTNNT2
HuTNNT2
6C2
6C2SRF
6C2cmv

Figure 5. Insertion of SRF binding element into chimeric CM promoter 6C2 did not alter the promoter strength. Replacing 100 bp proximal sequence with a 65 bp CMV core promoter sequence carrying TATA and TSS element increased the promoter strength by 2.4-fold compared to 400 bp hTNNT2 promoter.

TF binding rich regions of the TNNT2 promoters fused to CMV core promoter displayed promoter activity in iPSC-CM cells

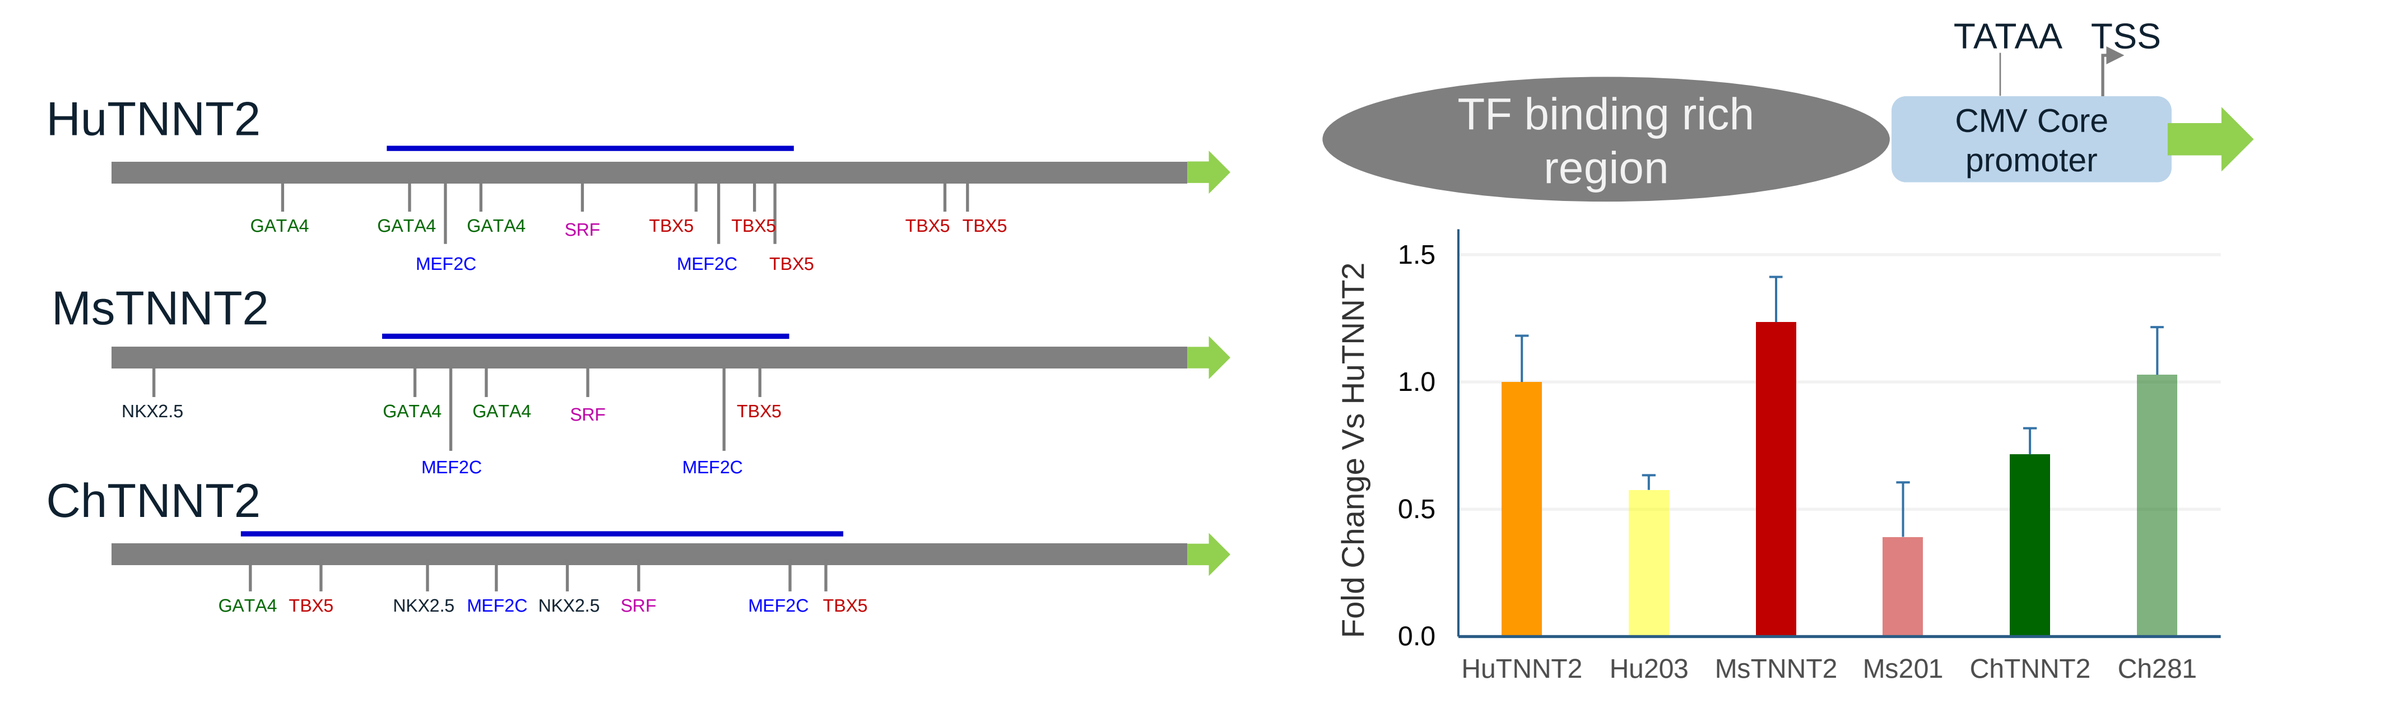
HuTNNT2
MsTNNT2
ChTNNT2
TF binding rich region
TATAA
TSS
CMV Core promoter
Fold Change Vs HuTNNT2
HuTNNT2
Hu203
MsTNNT2
Ms201
ChTNNT2
Ch281

Figure 6. CMmini promoters were constructed by fusing transcription factor binding rich regions (Solid blue bars) of 400 bp TNNT2 promoter from human (HuTNNT2), mouse (MsTNNT2) and chicken (ChTNNT2) with the 65 bp CMV core promoter. CMmini promoter derived from the human (Hu203) and mouse (Ms201) showed reduced promoter strengths compare to their respective parental promoter. The CMmini derived from the chicken (Ch281) exhibited enhanced (1.4-fold) promoter activity compared to its parental promoter.

RESULTS, cont'd

The modified 6C2 promoter exhibited equivalent gene expression levels in mouse heart, and enhanced liver expression compared to the HuTNNT2 promoter

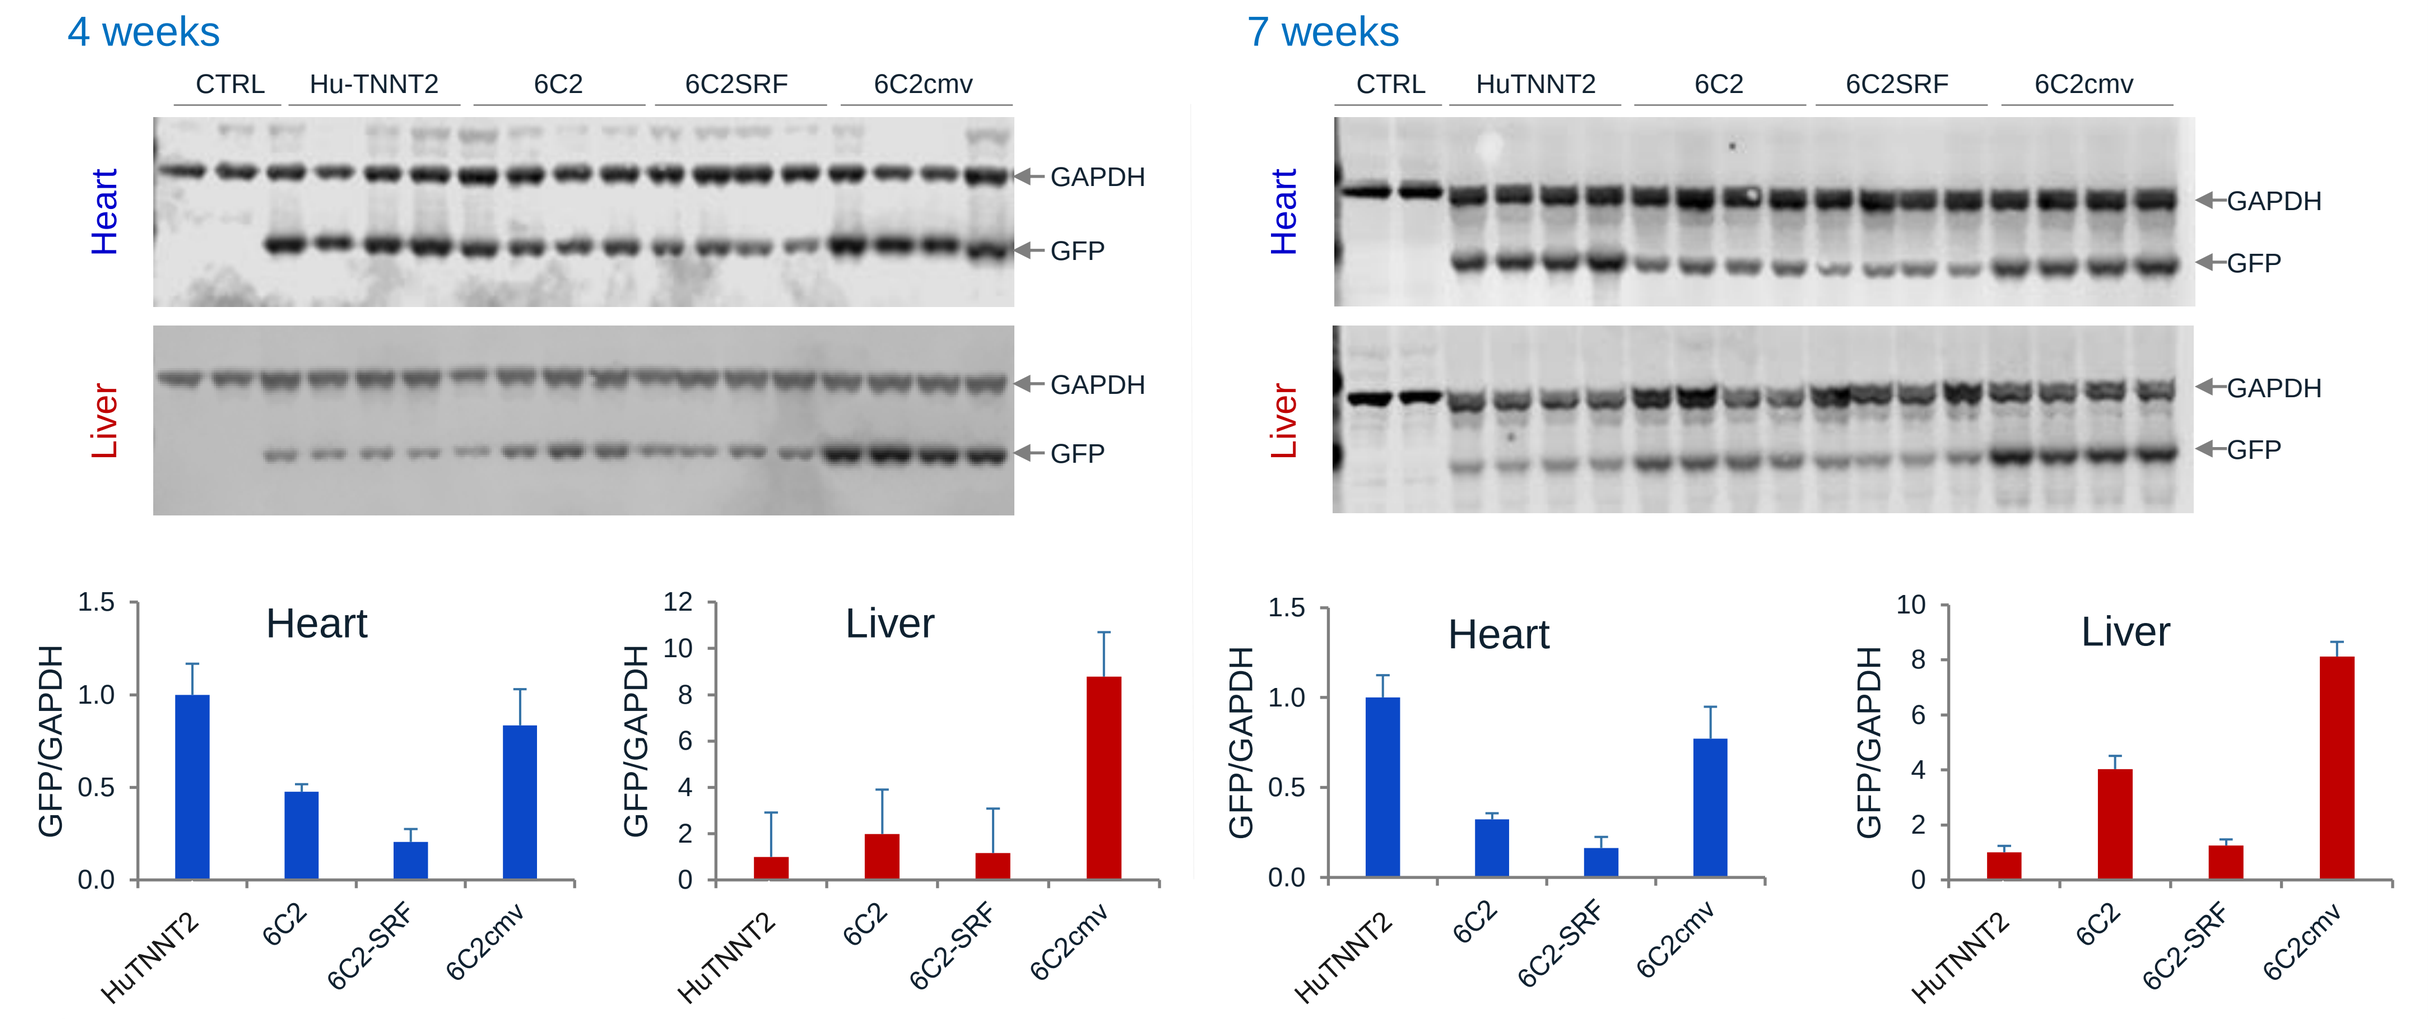
4 weeks
7 weeks
CTRL
Hu-TNNT2
6C2
6C2SRF
6C2cmv
Heart
Liver
GAPDH
GFP
GFP/GAPDH

Figure 7. Indicated promoter constructs directing the expression of GFP were packaged in AAV9 capsids and administered intravenously into mice (1E+13 vg/Kg body weight). 4 or 7 weeks later, promoter strengths were determined by immunoblotting for GFP in the hearts and livers.

Enhanced liver gene expression from the modified 6C2 promoter could be blunted by the addition of a single miR122 target sequence

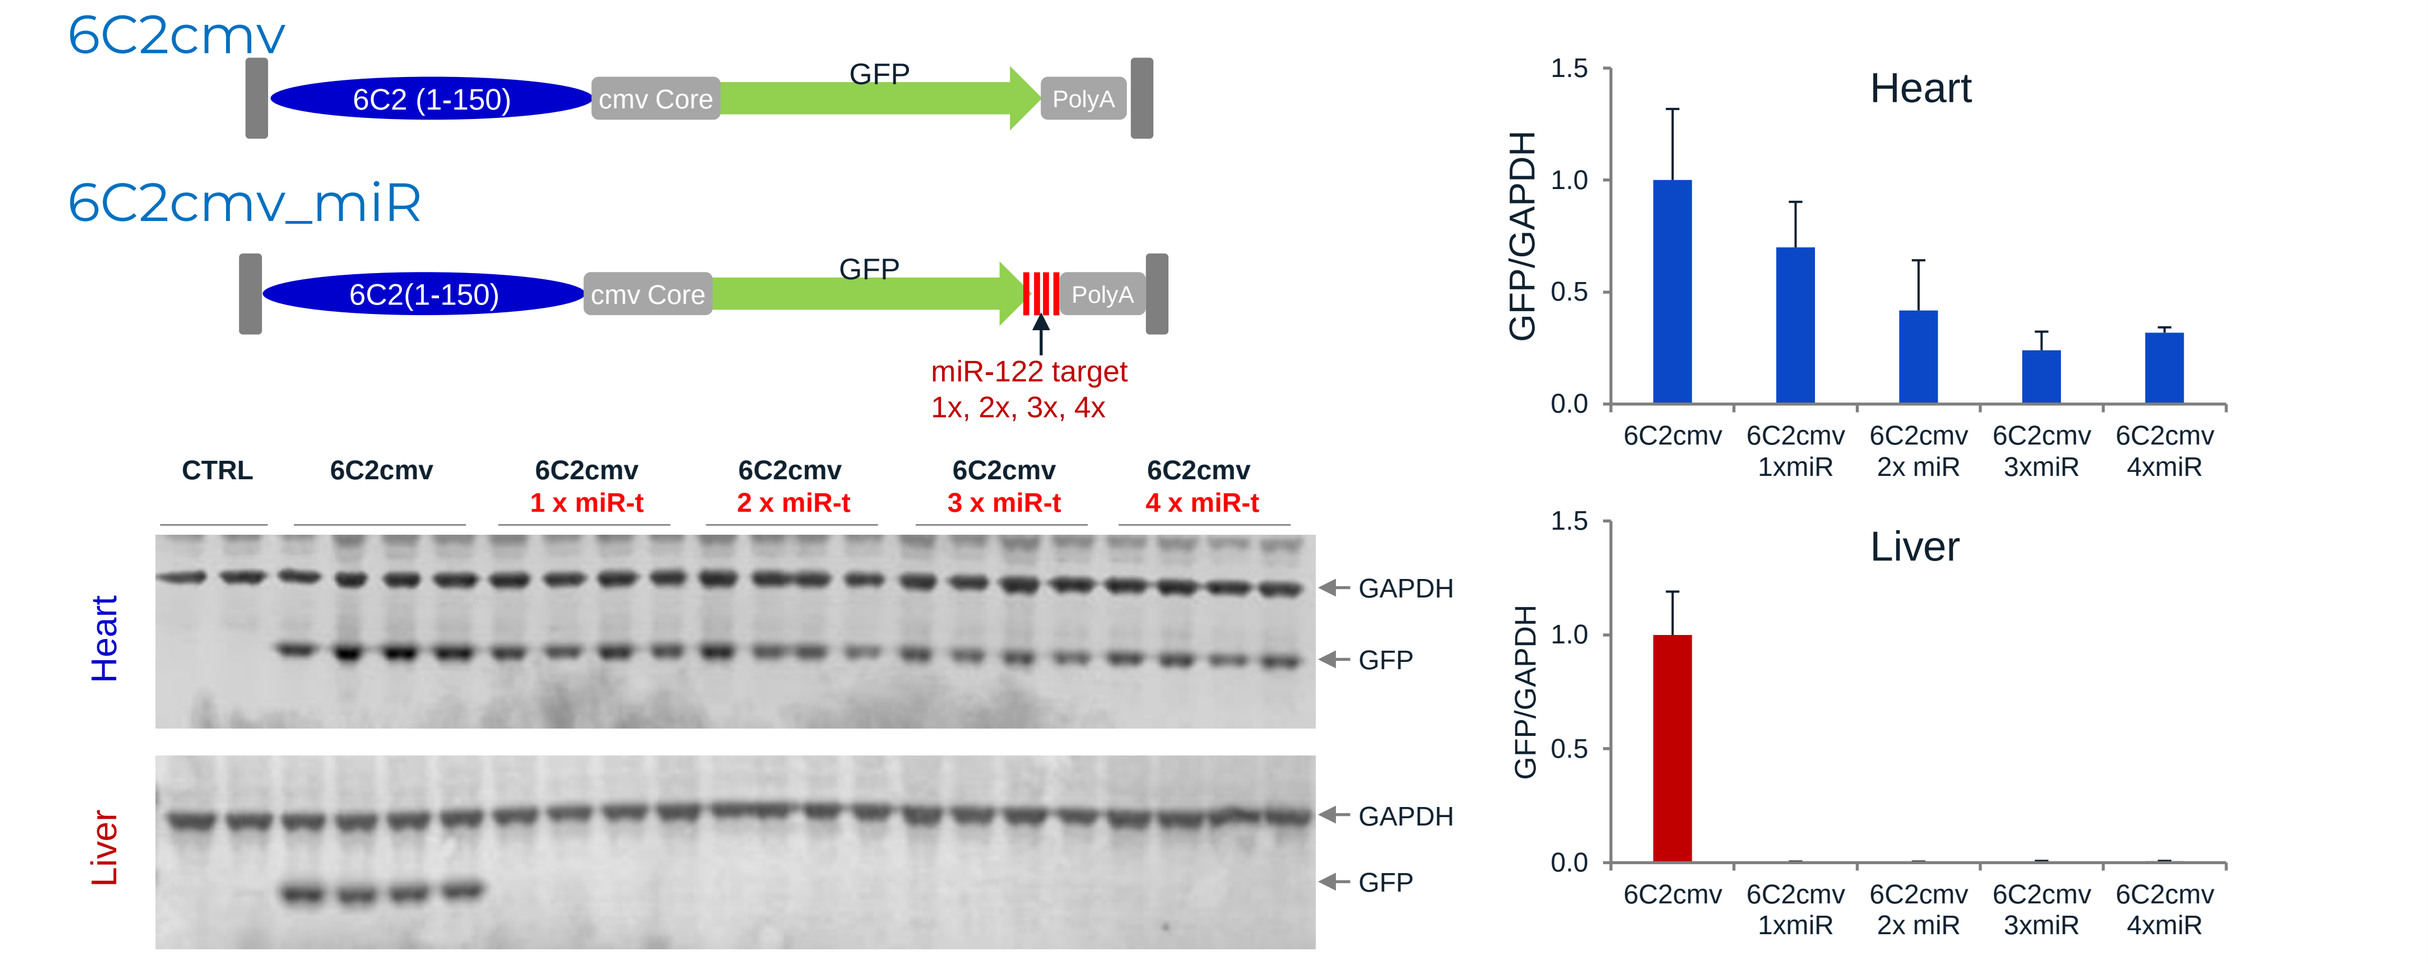
6C2cmv
6C2cmv_miR
6C2 (1-150)
cmv Core
GFP
PolyA
miR-122 target
1x, 2x, 3x, 4x
CTRL
6C2cmv
1 x miR-t
2 x miR-t
3 x miR-t
4 x miR-t
Heart
Liver
GAPDH
GFP
GFP/GAPDH

Figure 8. 6C2 promoter with tandem repeats of the indicated miR-122 target (miR-t) sequence located between the GFP ORF and the bGH PolyA signal were packaged in AAV9 capsids and administered by iv into mice. 4 weeks later, promoter strengths were determined by immunoblotting for GFP in the hearts and livers.

The modified 6C2 promoter together with bGH polyA exhibited superior long-term gene expression

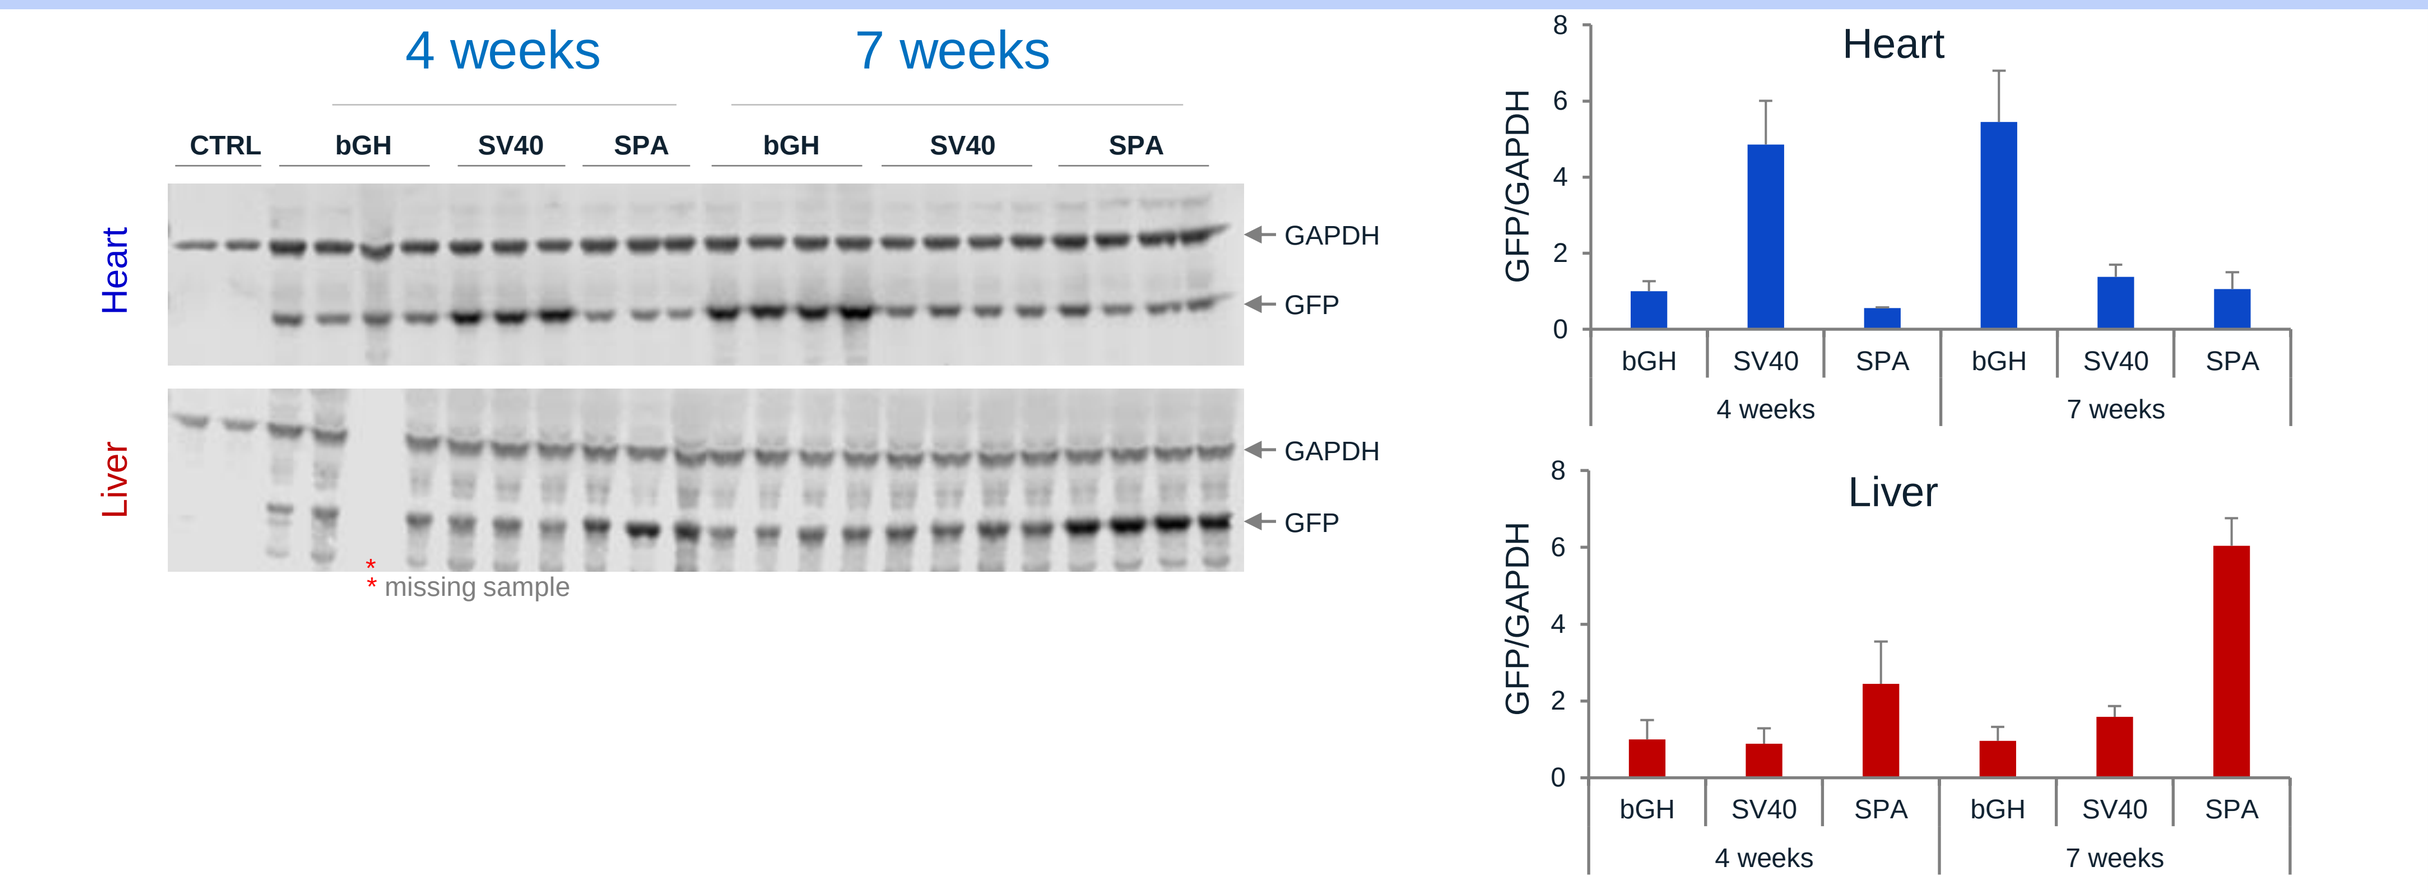
4 weeks
7 weeks
CTRL
bGH
SV40
SPA
Heart
Liver
GAPDH
GFP
* missing sample
GFP/GAPDH

Figure 9. Chimeric CM promoter 6C2cmv together with bGH, SV40 or SPA polyA signals directing the expression of GFP were packaged in AAV9 capsids and administered intravenously into mice (1E+13 vg/Kg body weight).). 4 or 7 weeks later, promoter strengths were determined by immunoblotting for GFP in the hearts and livers.

CONCLUSION

We have successfully engineered a compact 238 bp CM specific promoter, modified 6C2, exhibiting promoter strength comparable to that of the HuTNNT2 promoter

- Identified from a novel library of chimeric promoters constructed using the proximal promoter regions of the cardiomyocyte-specific genes.
- Modification of 6C2 via a) replacing proximal 100 bp region with the 65 bp CMV core promoter, b) addition of miR-122 target sequence downstream of the ORF, enhanced the promoter strength close to 400 bp human TNNT2 promoter, a gold standard for CM specific gene expression, and c) modified 6C2 together with bGH-polyA exhibited superior long term gene expression in the mouse heart.
- Further, validation of the modified 6C2 promoter's strength and specificity in large animal, and relevant disease model would be required for its utility in CM targeted gene therapy applications.

The novel, compact CM-specific promoter can facilitate cardiac gene therapy for larger genes of interest or cardiac gene editing allowing them to fit within the 4.7 kb capacity of AAV.