Development of a Scalable High Yield HEK293 Expression Platform for AAV Manufacturing

Chaz Feathers[1], Brooke Rathie[1], XiaoShan Ke[1], Jackson Leong[1], Joe Woods[1], Ze Cheng[1], Rafael Gamboa[1], Frank Jing[1], Bill Prince[1], Kee-Hong Kim[1]

[1]Tenaya Therapeutics, Inc. South San Francisco, CA – 94080, USA.

Introduction

For more than 20 years AAV therapeutics and technology have proven to be an efficient vehicle of delivery in academic and private sector cell and gene therapy. While many therapeutics have been proven safe and more recent BLA Approvals for commercialization confirm efficacy, the primary bottleneck to move the industry to the next phase of maturity is reduction in manufacturing cost while retaining safety, efficacy while significantly improving specific yields per Lot. Early approvals have continued to drive the interest in AAV technology for rare disease but therapeutic development into more prevalent disorders for gene replacement and gene regulation require significantly greater doses produced per Lot to meet potential demand of these indications and provide accessibly for the patients affected. While technology today cannot completely homogenize AAV production we believe steps can be taken to push AAV production toward a more desired and controlled product that can decouple this bottleneck in industrializing HEK293 transfection based AAV manufacturing.

In the pursuit of a universal novel platform, we considered elements contained in existing wildtype transfection-based expression systems that can be exploited. Publications such as Wörner et al. 2021, and Tran et al. 2022, have proven that AAV therapeutics made today are highly heterogeneous both in the stochastic nature of capsid assembly and the genome packaging variance of common recombinant AAVs, respectively. Primary measures of AAV production quality for both development and Drug Product release have historically relied explicitly on bulk measurements of capsid and genome quality. Current Expression systems regularly utilizing classic wildtype Cap and Rep constructs limit the potential of recombinant AAV expression technology without the incorporation of contemporary synthetic biology approaches.

To further address significant manufacturing costs developers today are additionally investing in dual plasmid transfection platform development (e.g. Tang et al. 2020; Lieshout et al. 2023; Velasquez et al. 2023), which possesses a clear advantage in starting material cost of plasmid manufacturing and streamlines complexity in cGMP transfection operations. These contemporary platform designs also add a potential advantage in product titer and % full capsids as only 2 plasmids must now transfect a cell to produce AAV therapeutics.

Objectives

- Design a library of Capsid, Rep and Helper constructs that can be used universally for any wildtype or novel capsid serotype
- Improve productivity of transfection based HEK293 suspension platforms by modernizing packaging components of AAV expression cassettes
- Reduce cost and transfection complexity with conversion of triple plasmid to a dual plasmid AAV expression system
- Scale Production of triple and dual plasmid formats into stir tank reactor demonstrating manufacturability
- Demonstrate that modifications made to HEK293 AAV expression system report comparable functional characterization by *in vitro* and *in vivo* product characterization

Material and Methods

- HEK and BEV libraries were first designed *in silico* and reviewed thoroughly prior to assembly of production and BEV transfer plasmids
- Plasmids were built in stages, when necessary, from PCR and synthesized DNA fragments
- Additional REP and Helper expression designs were also completed in this project for successful expression and recovery of AAV products
- Sf9 and HEK293 AAV manufacturing utilized Tenaya proprietary manufacturing platforms unless further disclosed throughout the presentation
 - The HEK293 production cell line utilized in this project, except when compared to Tenaya generated clones, is a commercially available clonal suspension cell line
- All Characterization work performed was completed at Tenaya by Tenaya PD and Translation Medicine Team
 - Western Blot, NAb Affinity, iPSC-CM transduction and ddPCR assays were developed by Tenaya
 - Capsid ELISA is performed with a commercially available kit

Icosahedral Rearrangement Technology (IRT)

Initial development and designs of Tenaya's Icosahedral Rearrangement Technology (IRT) were generated with the hypothesis that assembled capsid subunit protein ratios of the Sf9/BEV system could be systematically modified by a complete redesign of synthetic expression cassettes in a dual BEV platform. Sf9/BEV systems have endured a long history of sub optimal VP1 (Urabe et al. 2002) and VP2 (Chen 2008) ratios which could negatively affect the AAV capsid's target cell infectivity, endosomal escape, cytoplasmic trafficking and potentially therapeutic cassette transcription. The IRT expression system was intentionally designed for universal application to any serotype or novel capsid sequences. The specific control of the assembled capsid's subunit ratio applied to match or improve upon the canonical 1:1:10 (VP1:VP2:VP3), exploiting the stochastic nature of AAV capsid assembly by altering the composition of the available pool of unassembled subunit proteins during AAV production.

Figure 1. Wörner et al. 2021

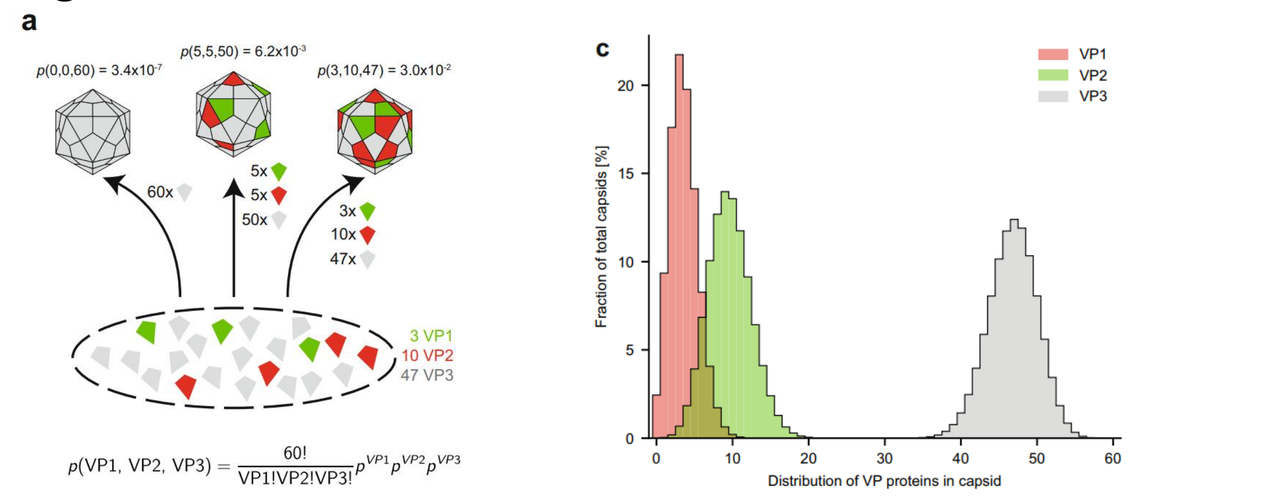
a
c
60x
3 VP1
10 VP2
47 VP3
VP1
VP2
VP3
Fraction of total capsids [%]
Distribution of VP proteins in capsid

Figure 1. **a** Illustration of AAV capsid assembly following a stochastic model, where VPs are drawn randomly from the pool of expressed VPs. The probability for each capsid stoichiometry is solely determined by the ratio of the expressed VPs. **c** Summed probabilities for capsids having a specific number of VP1, VP2, or VP3 in their capsid.

Figure 2.

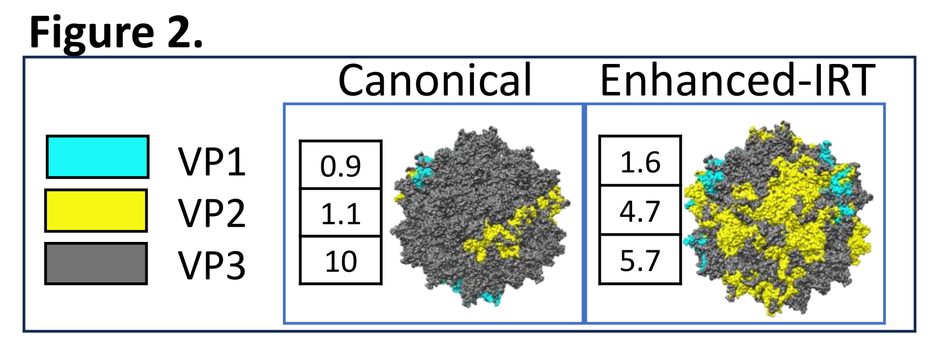
Canonical
Enhanced-IRT
VP1
VP2
VP3
0.9
1.1
10
1.6
4.7
5.7

Figure 2. Superimposed cartoon representation of Enhanced VP1 and VP2 subunits with a 3-D, AAV9-IRT capsid model.

Figure 3a. **Western Blot of Affinity Purified Sf9-IRT AAV9**

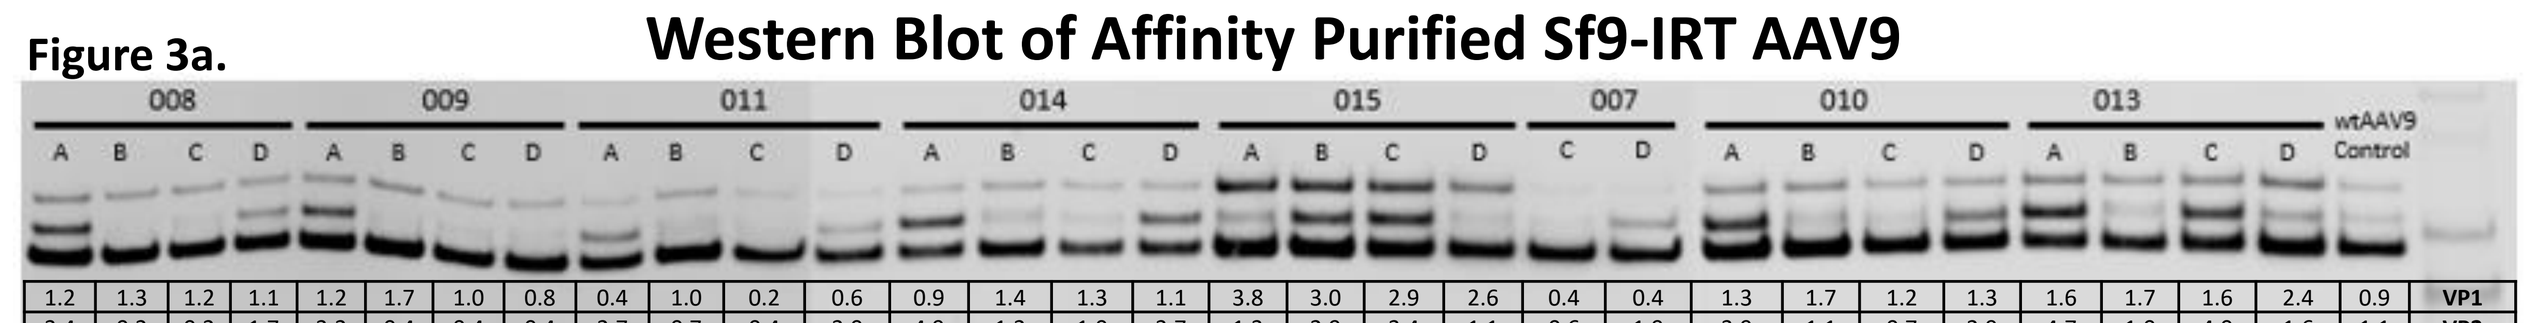
008
009
011
014
015
007
010
013
wtAAV9 Control
VP1
VP2
VP3

	008 A	008 B	008 C	008 D	009 A	009 B	009 C	009 D	011 A	011 B	011 C	011 D	014 A	014 B	014 C	014 D	015 A	015 B	015 C	015 D	007 C	007 D	010 A	010 B	010 C	010 D	013 A	013 B	013 C	013 D	wtAAV9 Control
VP1	1.2	1.3	1.2	1.1	1.2	1.7	1.0	0.8	0.4	1.0	0.2	0.6	0.9	1.4	1.3	1.1	3.8	3.0	2.9	2.6	0.4	0.4	1.3	1.7	1.2	1.3	1.6	1.7	1.6	2.4	0.9
VP2	3.4	0.2	0.2	1.7	3.2	0.4	0.4	0.4	2.7	0.7	0.4	2.0	4.9	1.2	1.0	3.7	1.3	2.9	3.4	1.1	0.6	1.9	3.9	1.1	0.7	2.9	4.7	1.0	4.0	1.6	1.1
VP3	7.4	10.5	10.6	9.2	7.5	9.9	10.6	10.8	8.9	10.3	11.4	9.4	6.3	9.5	9.7	7.2	7.0	6.0	5.7	8.4	11.0	9.7	6.8	9.2	10.1	7.8	5.7	9.2	6.4	8.1	10.0

Figure 3b.

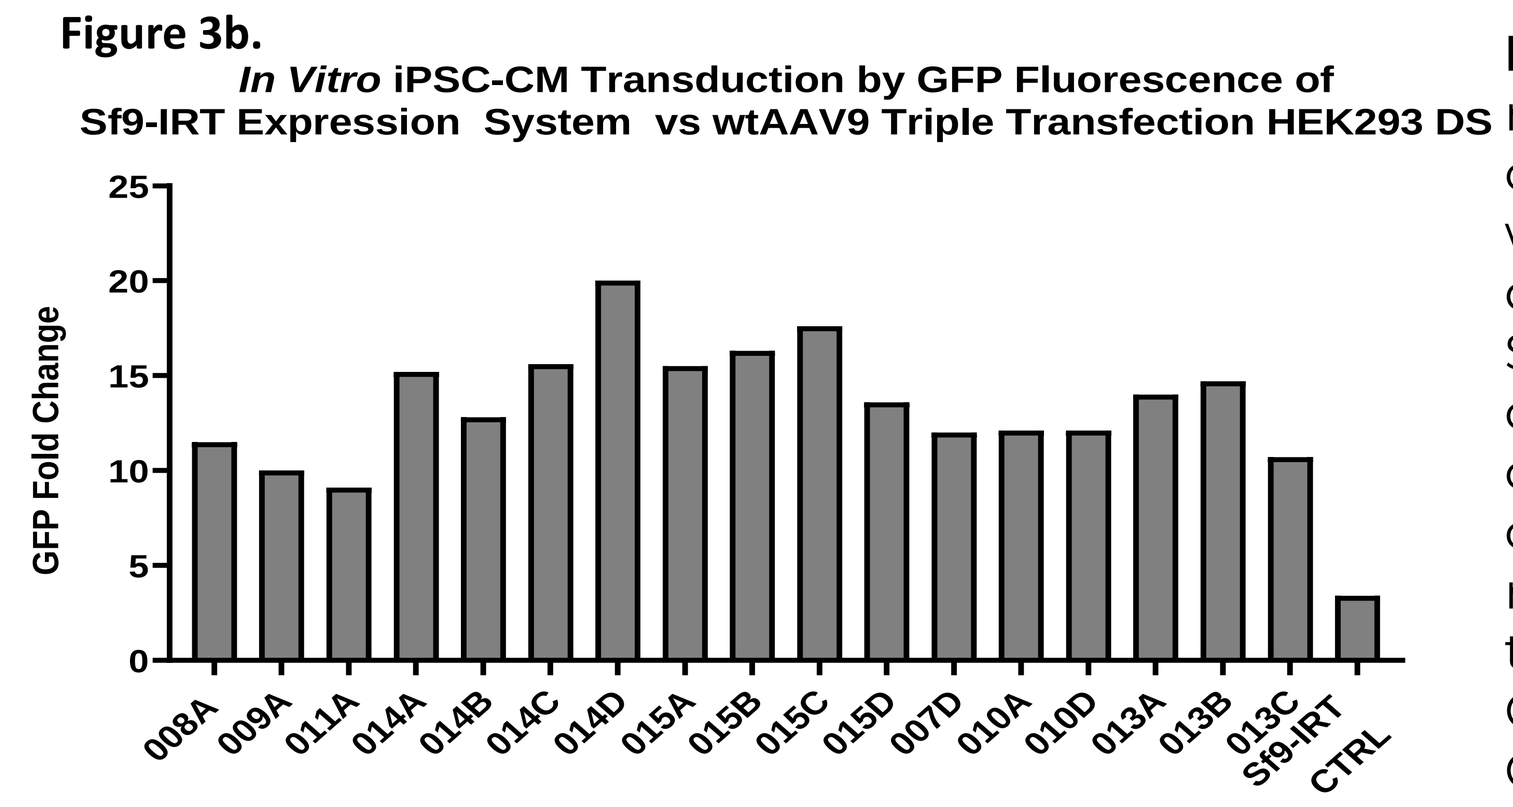
In Vitro iPSC-CM Transduction by GFP Fluorescence of
Sf9-IRT Expression System vs wtAAV9 Triple Transfection HEK293 DS
GFP Fold Change
008A
009A
011A
014A
014B
014C
014D
015A
015B
015C
015D
007D
010A
010D
013A
013B
013C
Sf9-IRT CTRL

Figure 3. **a** Western blot of manufacturable IRT diversity post affinity capture purification. Sf9-IRT constructs were first tittered by ddPCR which demonstrated comparable titers to classic Sf9 system then further purified for characterization **b** *in vitro* iPSC-cardiomyocyte (CM) transduction assay observes that altering assembled capsid ratios confers significant advantage in transduction reported through translated GFP fluorescence. Imaging performed on Cytation imager.

IRT-AAV9 Conversion for Mammalian Expression

Productivity is a significant bottleneck in HEK293 AAV production so borrowing from our high yield novel IRT Sf9/BEV system we converted IRT constructs for compatibility with mammalian expression platforms. In this effort we targeted a range of constructs predicted to produce assembled capsid subunit stoichiometry in a range of the canonical 1:1:10 ratio. Here we demonstrated numerous iterations of IRT capsid subunit protein control that are comparable to classic wtAAV9 expression systems. Many produce comparable titer to classic wtAAV expression platforms. One construct, 17D, was pursued for further interrogation due to both it's ability to generate canonical capsid subunit ratios and significant improvement in productivity.

All products characterized by western blot below have been purified by affinity capture further demonstrating that IRT constructs produce stable AAV products.

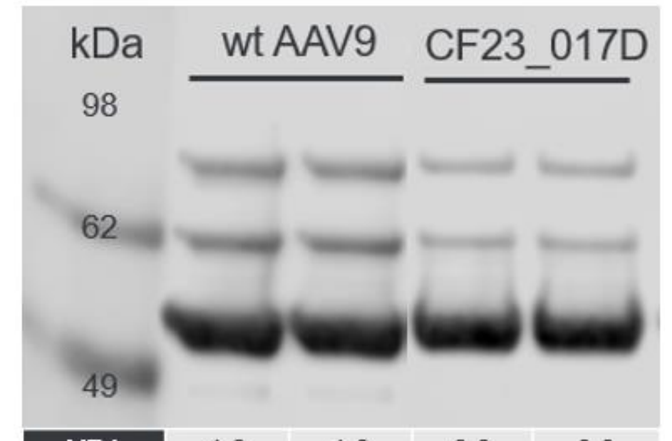
kDa
wt AAV9
CF23_017D
98
62
49

	wt AAV9	wt AAV9	CF23_017D	CF23_017D
VP1	1.8	1.8	0.9	0.9
VP2	2.4	2.4	1.0	1.0
VP3	7.8	7.8	10.1	10.1

Figure 4a. Western Blot overloaded comparing affinity captured wtAAV9 to High yield 17D-IRT.

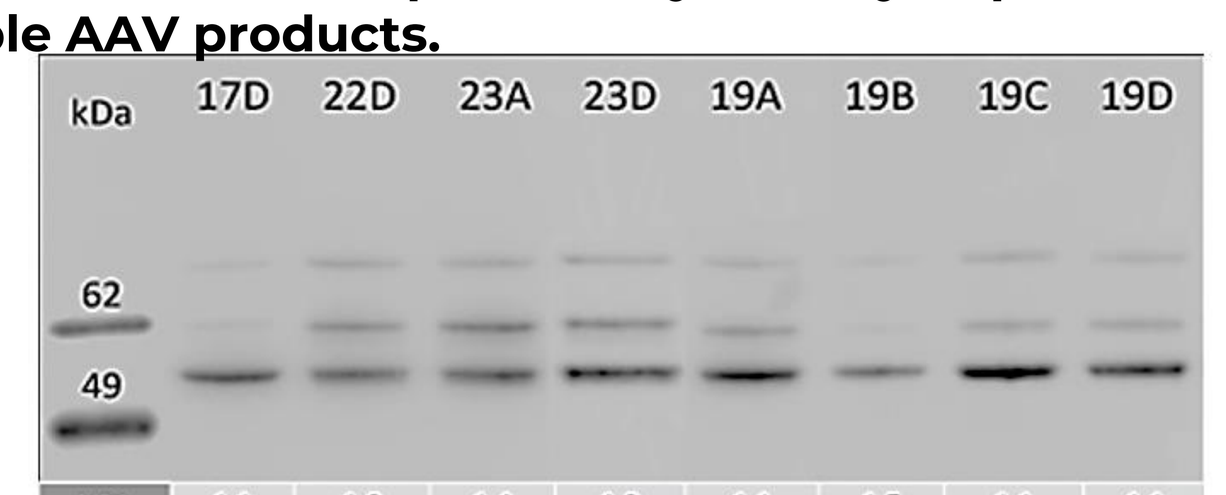
kDa
17D
22D
23A
23D
19A
19B
19C
19D
62
49

	17D	22D	23A	23D	19A	19B	19C	19D
VP1	1.1	1.8	1.4	1.2	1.1	1.5	1.1	1.1
VP2	1.2	3.6	4.5	3.3	2.5	1.4	1.5	1.9
VP3	9.8	6.7	6.2	7.5	8.4	9.1	9.4	9.0

Figure 4b. Additional Constructs from Tenaya's IRT Library with variable capsid subunits which post affinity capture have assembled capsids comparable to wtAAV. Some variability is observed in the amount of product loaded onto SDS-PAGE.

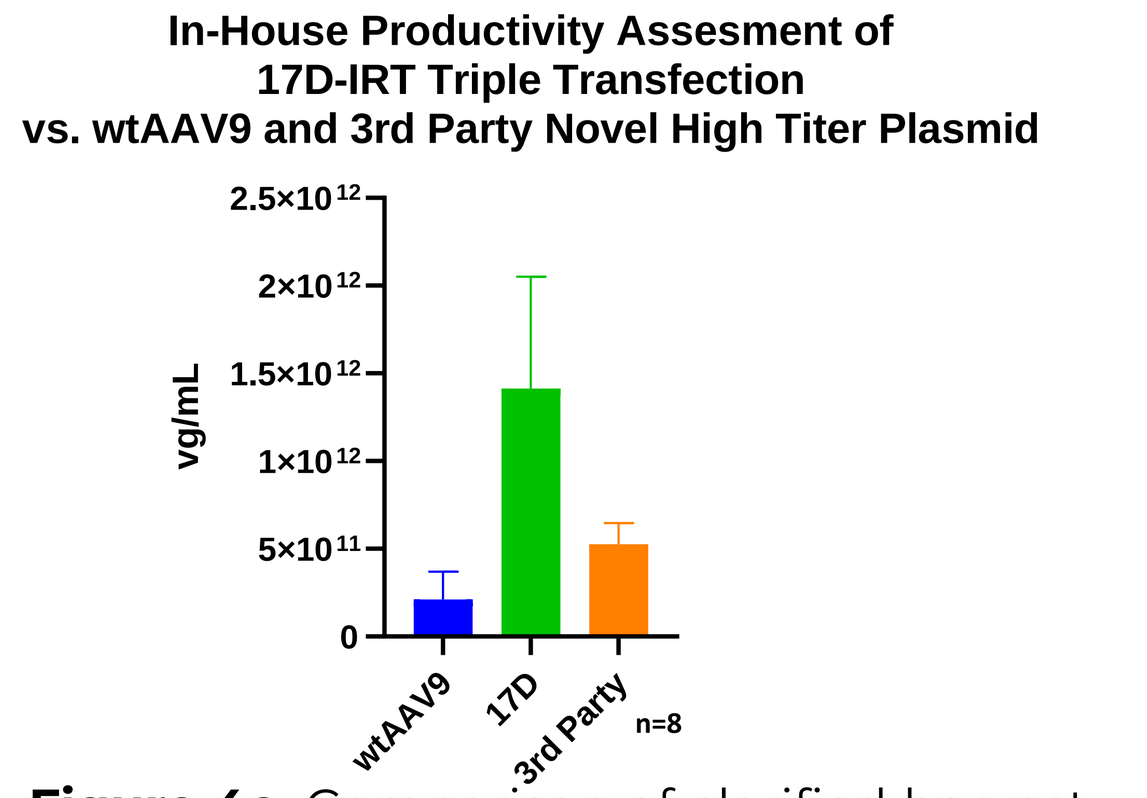
In-House Productivity Assesment of
17D-IRT Triple Transfection
vs. wtAAV9 and 3rd Party Novel High Titer Plasmid
vg/mL
wtAAV9
17D
3rd Party
n=8

Figure 4c. Comparison of clarified harvest titers over multiple shake flask productions demonstrating construct 17D has superior productivity to wtAAV and a 3rd party novel expression system designed for yield improvement.

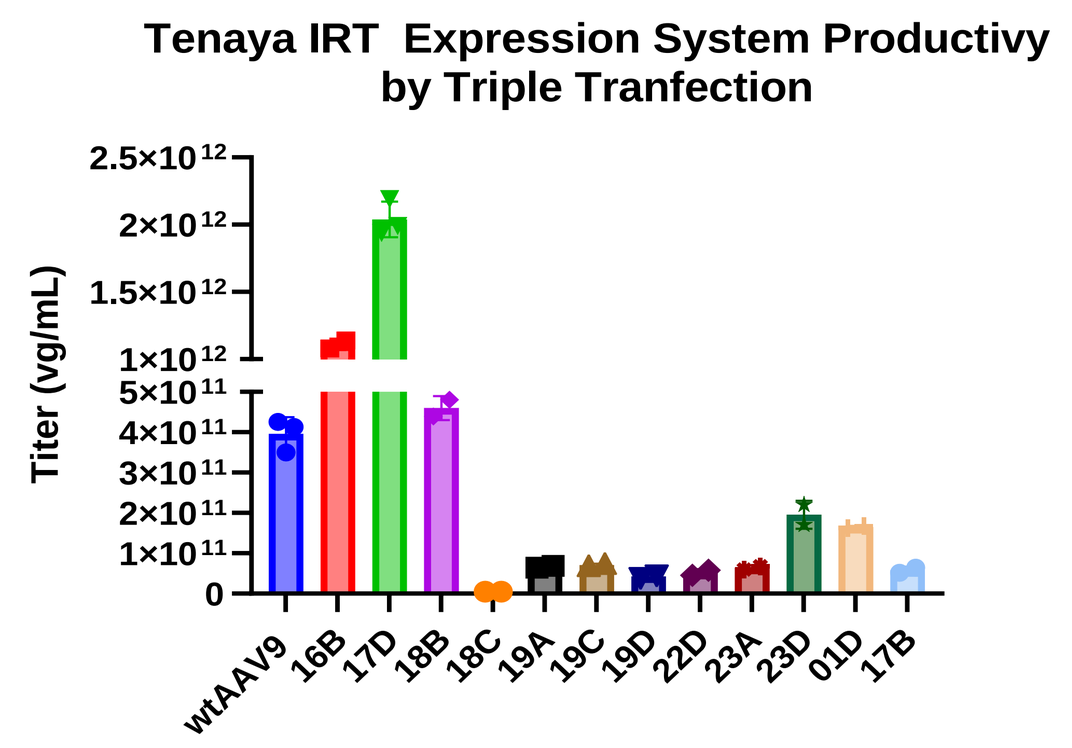
Tenaya IRT Expression System Productivy
by Triple Tranfection
Titer (vg/mL)
wtAAV9
16B
17D
18B
18C
19A
19C
19D
22D
23A
23D
01D
17B

Figure 4d. Clarified Harvest titers for a selection of Tenaya's IRT library which produce assembled capsids with compositions comparable to wtAAV.

Scale-Up of IRT Triple and Dual AAV Expression System

Improvements to reduce manufacturing cost were made in reformatting the IRT and wildtype triple plasmid transfection expression system to a dual plasmid format. Reformatting of classic wildtype or IRT system did not have an impact in shake flask productivity experiments performed. To demonstrate scale independence of the 17D-IRT construct both triple and dual expression systems were scaled to 50L Stir tank bioreactors in a head-to-head study.

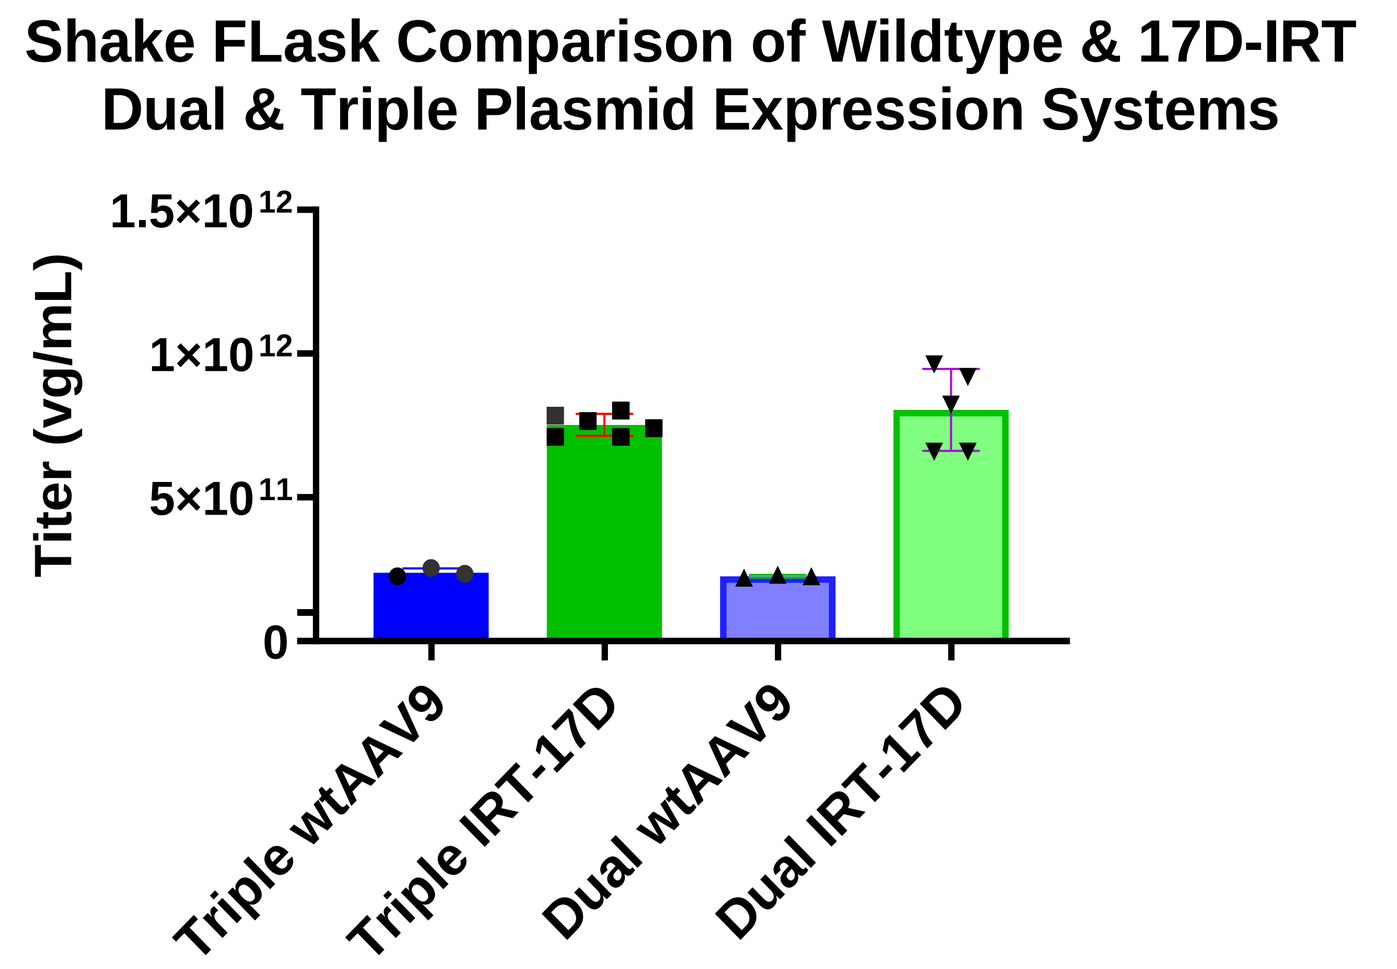
Shake FLask Comparison of Wildtype & 17D-IRT
Dual & Triple Plasmid Expression Systems
Titer (vg/mL)
Triple wtAAV9
Triple IRT-17D
Dual wtAAV9
Dual IRT-17D

Figure 5. Early dual plasmid system experimentation in shake flask productivity for wildtype and 17D-IRT demonstrated no difference in clarified harvest titer in dual transfection expression platform when produced head-to-head to their original triple plasmid format.

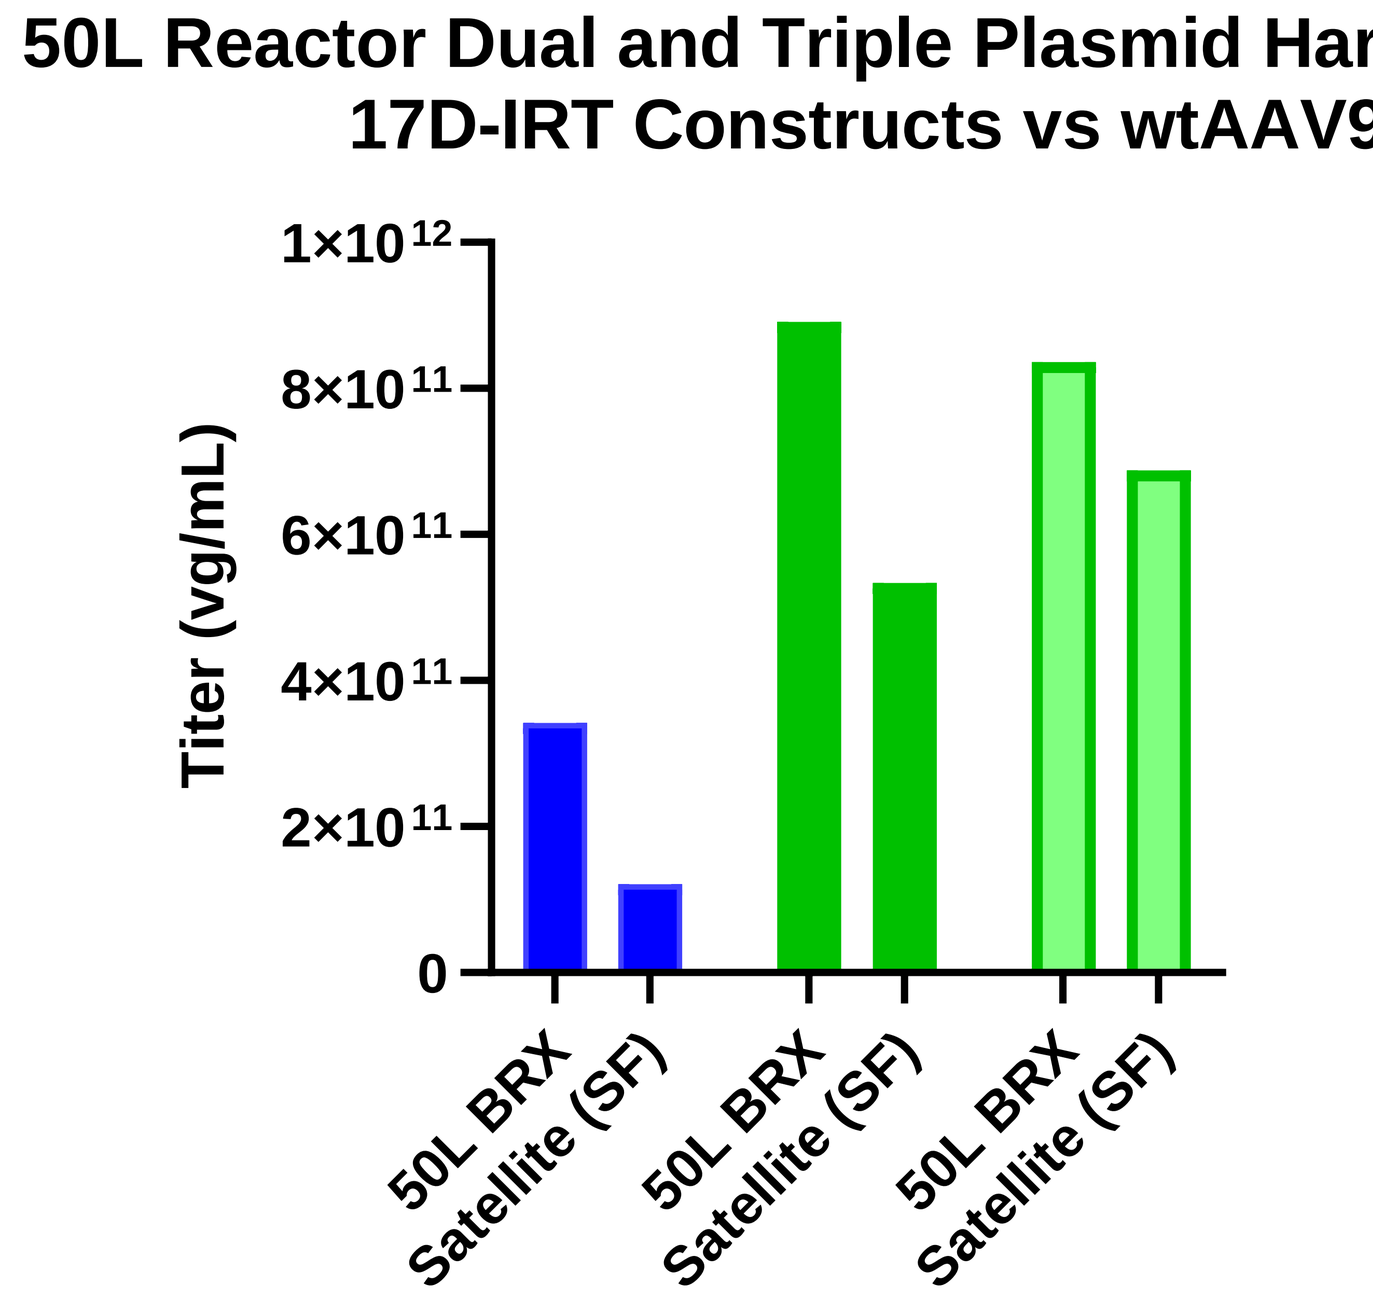
50L Reactor Dual and Triple Plasmid Harvest Titer
17D-IRT Constructs vs wtAAV9
Titer (vg/mL)
50L BRX
Satellite (SF)
wtAAV9 Triple TFX
17D-IRT Triple TFX
17D-IRT Dual TFX

Figure 6. 50L Scale up of construct 17D-IRT reported significant productivity increase >2.5-fold over wtAAV9 while retaining comparable titer in either triple of dual plasmid format expression system as previously observed in shake flask experiments. Dual plasmid 17D additionally offers a significant advantage in manufacturing cost and streamlines transfection complex formations in that the titer is comparable to triple plasmid system and reduces plasmid manufacturing requirement by 1/3.

In Vitro Comparability Characterization

After demonstrating through multiple productivity experiments the superior yield observed from IRT construct 17D an *in vivo* expression platform and producer cell line, (HEK293 suspension (Susp) or HEK293T adherent), comparability study was performed in healthy mice (Study not completed at time of presentation). This study is comparing total GFP translation in cardiomyocytes 3 weeks post injection. All products were first characterized *in vitro* for capsid composition, % full by ddPCR/ELISA (vg/cp), neutralizing antibody binding affinity (NAb) and GFP translation in human derived iPSC cardiomyocytes (iPSC-CM).

Table1.

	HEK293 Suspension Classic Triple Plasmid	HEK293T Classic Triple Plasmid	HEK293 Suspension IRT Triple Plasmid
Purpose	wtAAV9 Study Control	wtAAV9 Cell Line Comparability	High Titer Novel System
Construct	wtAAV9:GFP	wtAAV9:GFP	17D-IRT AAV9:GFP
Purification Method	IDX	IDX	IDX
Full Ratio% (vg/cp)	45%	40%	46%

7a.

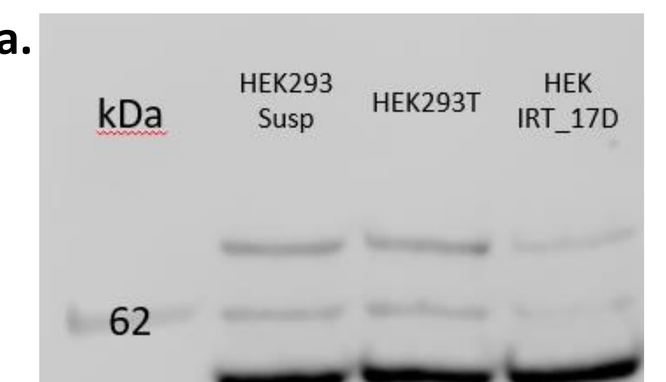
kDa
HEK293 Susp
HEK293T
HEK IRT_17D
62
49

	HEK293 Susp	HEK293T	HEK IRT_17D
VP1	1.5	1.6	0.7
VP2	1.2	1.2	0.5
VP3	9.3	9.3	10.8

7b.

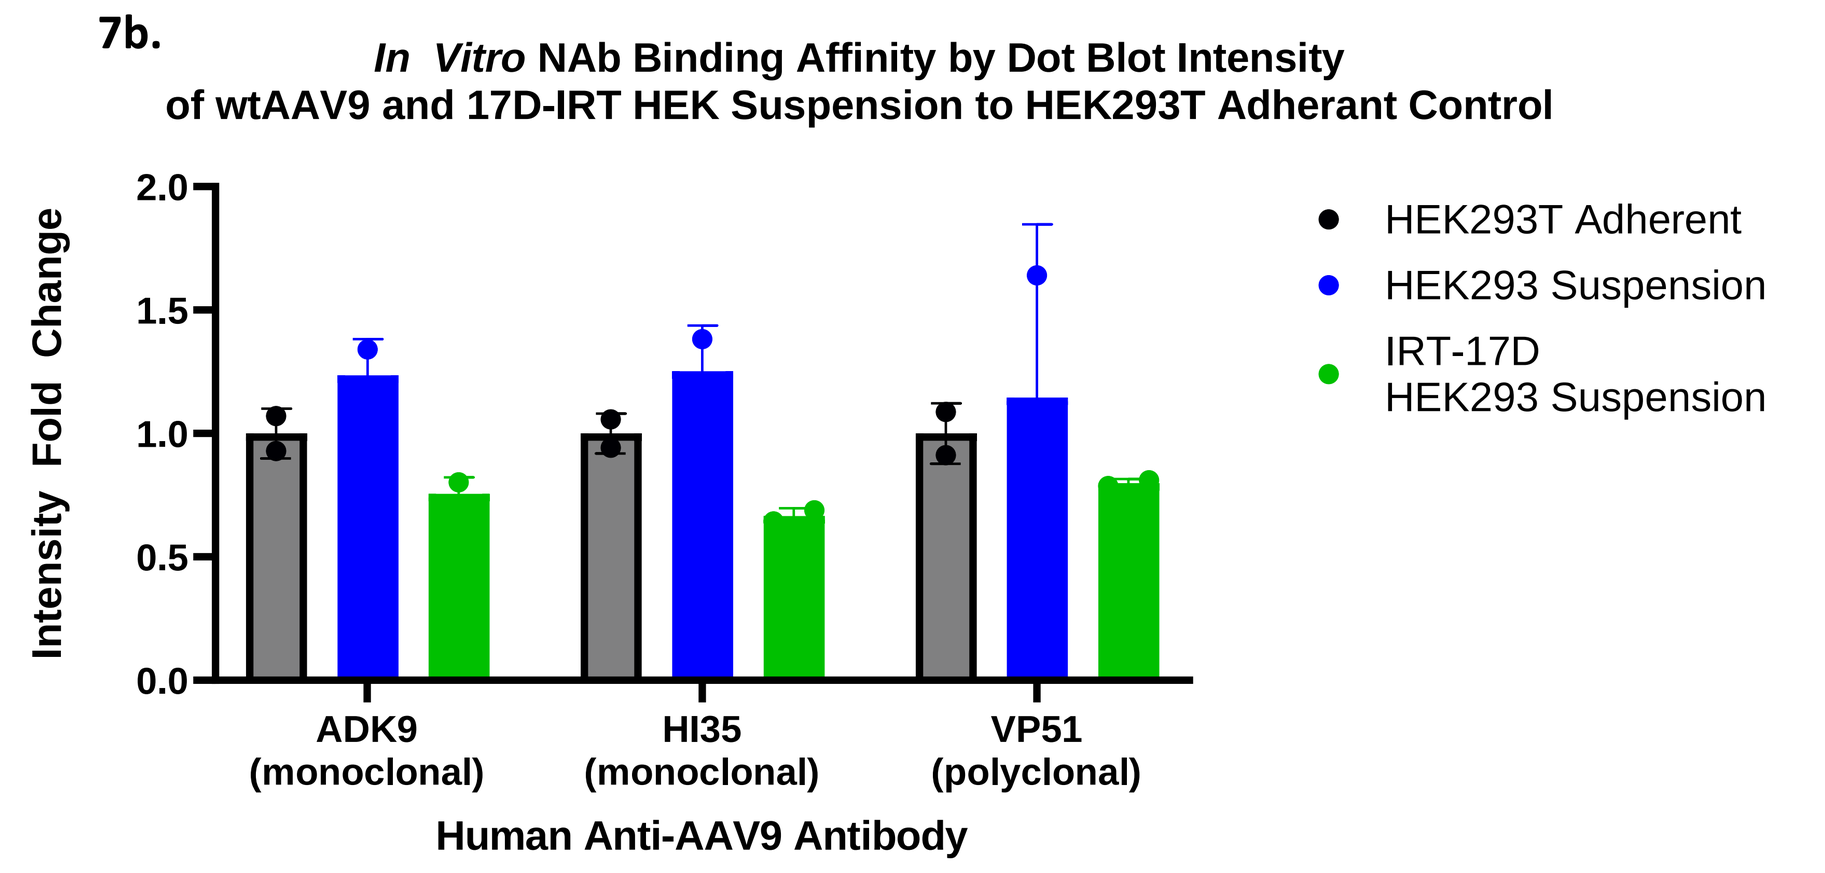
In Vitro NAb Binding Affinity by Dot Blot Intensity
of wtAAV9 and 17D-IRT HEK Suspension to HEK293T Adherant Control
Intensity Fold Change
ADK9 (monoclonal)
HI35 (monoclonal)
VP51 (polyclonal)
Human Anti-AAV9 Antibody
HEK293T Adherent
HEK293 Suspension
IRT-17D HEK293 Suspension

7c.

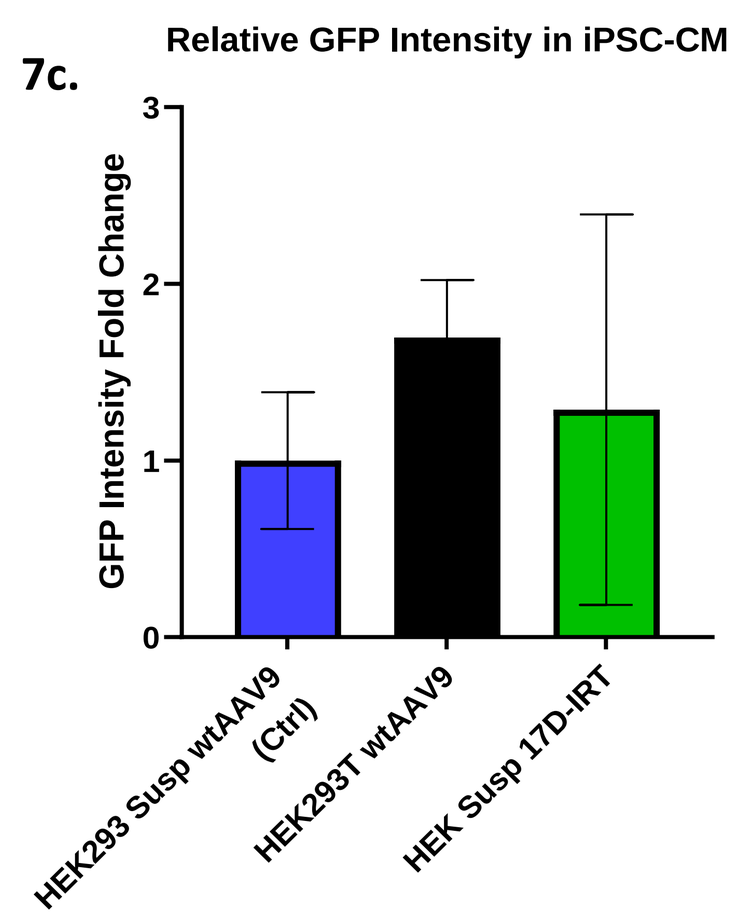
Relative GFP Intensity in iPSC-CM
GFP Intensity Fold Change
HEK293 Susp wtAAV9 (Ctrl)
HEK293T wtAAV9
HEK Susp 17D-IRT

Table 1. AVV9 products were produced by triple transfection in respective cell lines indicated and purified by Iodixanol (IDX) to maintain comparability. All products observed comparable % full ratio by ELISA/ddPCR

Figure 7a,b,c. AAV Products demonstrated comparable *in vitro* characterization prior to injections

HEK293 Clonal Cell Line Comparability

To further investigate construct 17D-IRT productivity both wtAAV9 and 17D-IRT were tested in a head-to-head productivity comparability of Tenaya derived HEK293 clones and a commercially available cell line. All productions were in shake flask with triple plasmid production of wtAAV9:GFP or 17D-IRT:GFP. Clarified harvest titers reported comparable wtAAV9 titers for clones C6 and C20 to that of the commercially available cell line. Construct 17D-IRT proved transferable in enhanced productivity between clonal cell lines and the commercially available cell line also used for previous productions presented (**Figure 8)**.

Clonal cell Lines were derived from ATCC1573.3 bank. The parental bank used in clonal development had previously been suspension and FBS free adapted in chemically defined medium. Clones selected for expansion and testing were again adapted to FBS free suspension in chemically defined medium after limited dilution plating. Clonal cell lines developed by Tenaya PD demonstrated high viability throughout expansion and re adaption with stabilized population doubling times **(Figure 9a & 9b)**.

Figure 8.

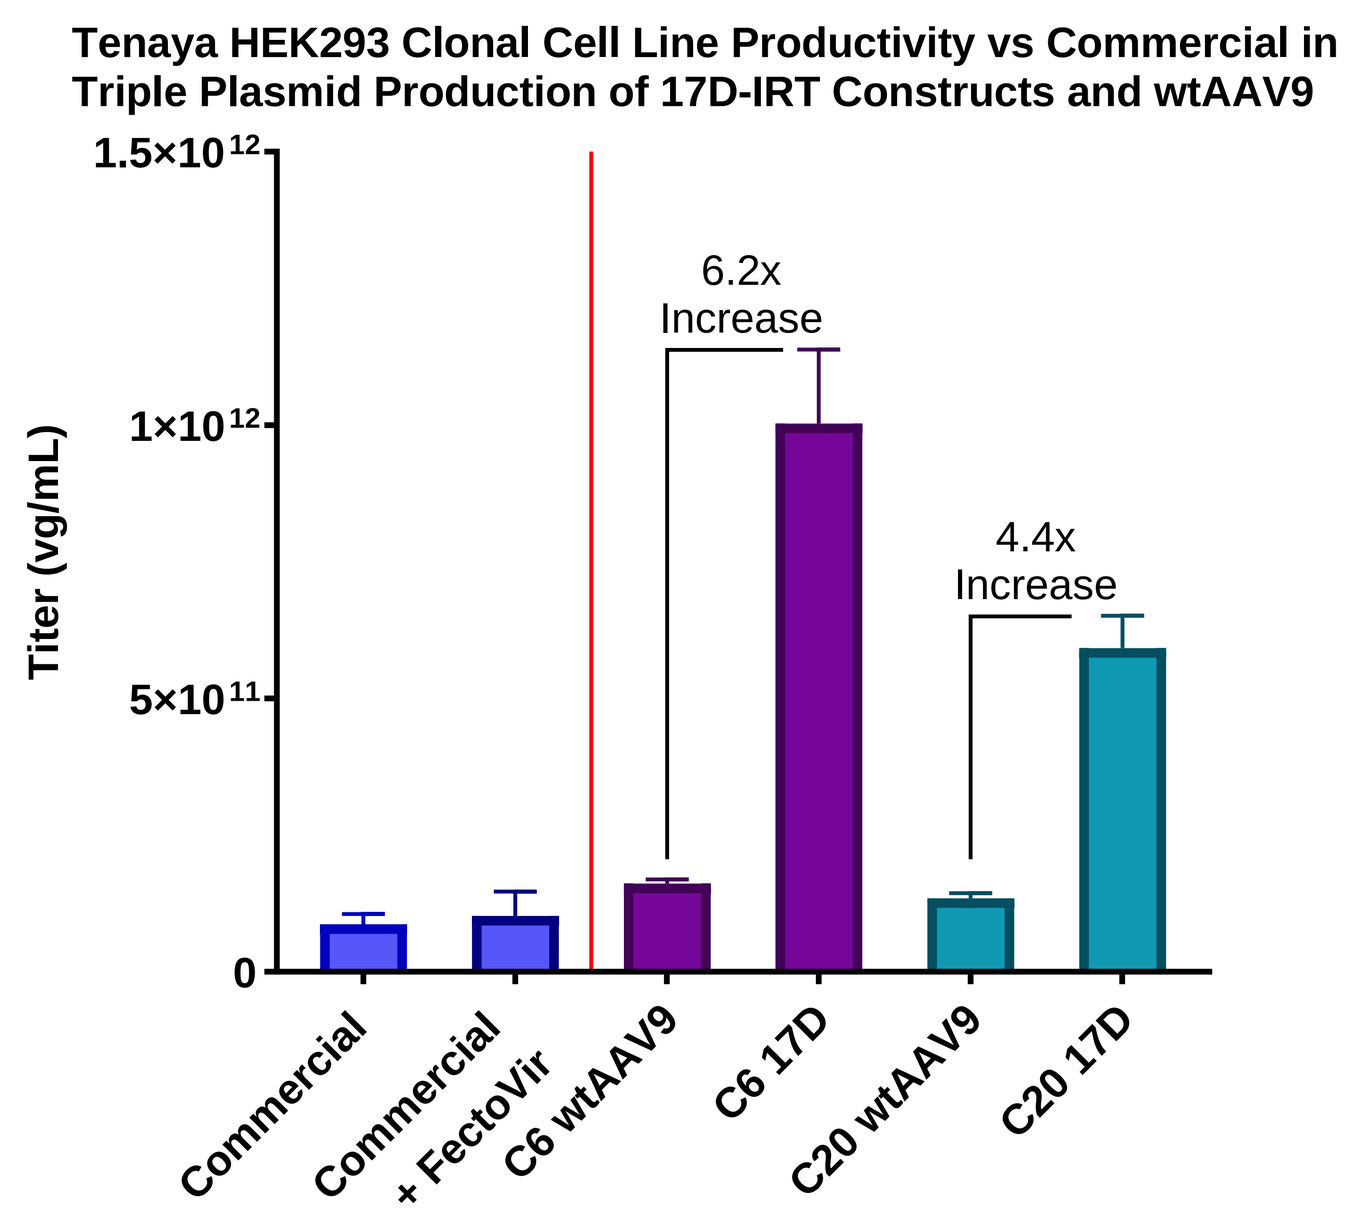
Tenaya HEK293 Clonal Cell Line Productivity vs Commercial in
Triple Plasmid Production of 17D-IRT Constructs and wtAAV9
Titer (vg/mL)
6.2x Increase
4.4x Increase
Commercial
Commercial + FectoVir
C6 wtAAV9
C6 17D
C20 wtAAV9
C20 17D

9a.

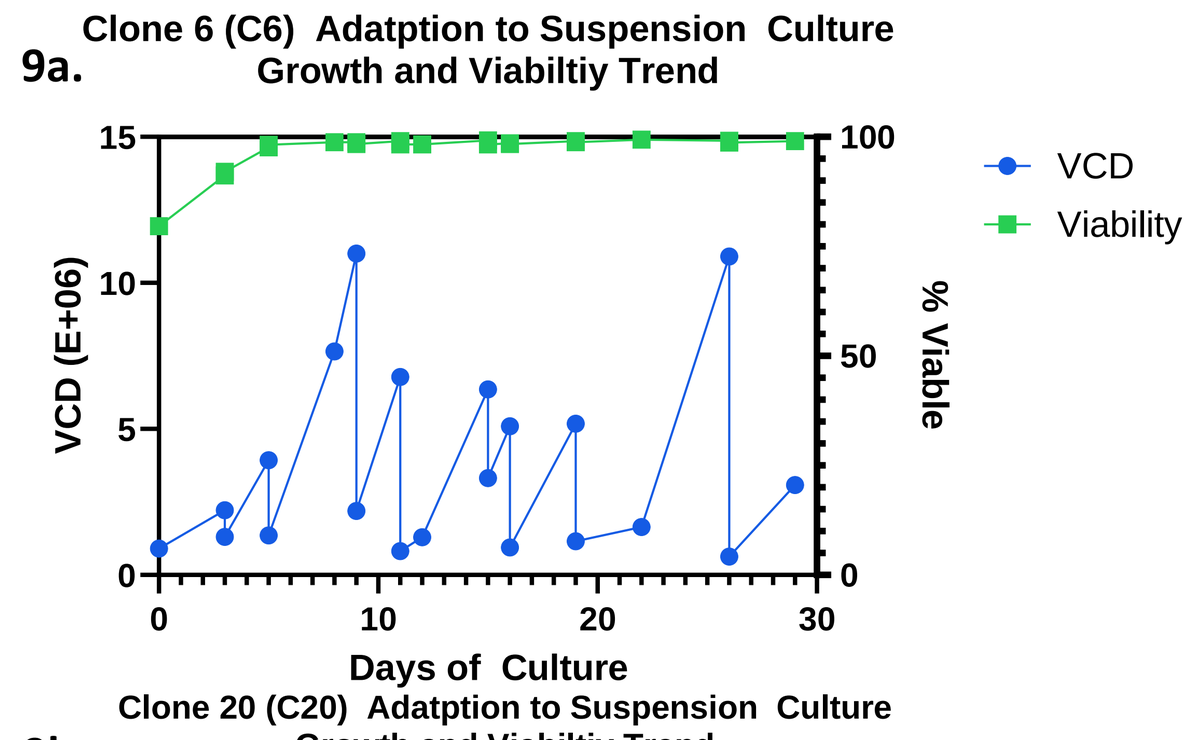
Clone 6 (C6) Adatption to Suspension Culture
Growth and Viabiltiy Trend
VCD (E+06)
% Viable
Days of Culture
VCD
Viability

9b.

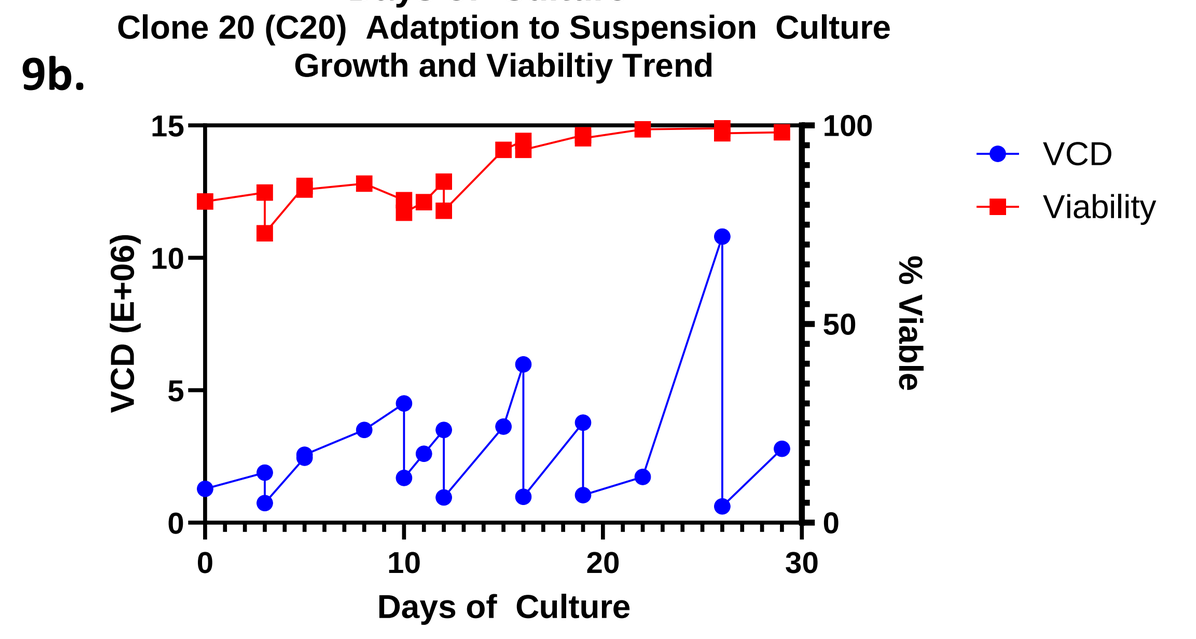
Clone 20 (C20) Adatption to Suspension Culture
Growth and Viabiltiy Trend
VCD (E+06)
% Viable
Days of Culture
VCD
Viability

Conclusions

- Successfully generated library of constructs with universal serotype application (IRT) via specific control of capsid subunit translation allowing for modulation of capsid subunit ratios in assembled capsids. A portion of these capsids recreated for HEK293 platform generate wildtype AAV capsid stoichiometry observed in classic recombinant expression systems
- Construct 17D-IRT with canonical capsid subunit ratios has robustly demonstrated significantly greater productivity than wildtype AAV9 and 3rd party novel systems pushing closer to meeting the high demand for upstream AAV yields and providing more patient doses per production Lot
- Dual and triple plasmid systems for 17D-IRT have been proven scalable with comparable productivity through 50L Stir Tank Reactor creating potential for further reduction in manufacturing costs of HEK293-AAV
- Additional Characterization of 17D products *in vitro* demonstrates comparability in % full capsids, iPSC-CM transduction and NAb binding affinity making it a suitable candidate for *in vivo* Studies
- Additional productivity experiments performed in two clonal HEK293 cell lines developed by Tenaya PD demonstrate consistent productivity of wtAAV9 to a commercial clonal cell line and transferable enhanced productivity when construct 17D-IRT is utilized.

Acknowledgements

Thank you to all Tenaya collaborators.

- Gene Therapy Research and Translational Medicine Teams for their execution of the *in vivo* experiment and reporting as well as NAb characterization
- Process Development Team where construct builds, screening productions, many characterization assays and scale-up took place

Contact Chaz Feathers, cfeathers@tenayathera.com